339

ADXS11-001 immunotherapy in squamous or non-squamous persistent/recurrent metastatic cervical cancer: Results from stage 1 [and stage 2] of the phase II GOG/NRG-0265 study

Authors: Warner Huh, MD[1], Don Dizon, MD[2], Matthew A. Powell, MD[3], Charles A. Leath III, MD[1], Lisa M. Landrum, MD[4], Edward Tanner, MD[5], Robert Higgins, MD[6], Stefanie Ueda, MD[7], Michael McHale, MD[8], Bradley J. Monk, MD[9], Carol Aghajanian, MD[10]

Affiliations: [1]University of Alabama at Birmingham, Obstetrics/Gynecology, Birmingham, AL; [2]Massachusetts General Hospital Cancer Center, Gynecologic Oncology, Boston, MA; [3]Washington University School of Medicine, Obstetrics/Gynecology, St. Louis, MO; [4]University of Oklahoma Health Sciences Center, Section of Gynecologic Oncology, Oklahoma City, OK; [5]Johns Hopkins Medical Institutions, Gynecology and Obstetrics, Baltimore, MD; [6]Carolinas Medical Center, Obstetrics/Gynecology, Charlotte, NC; [7]UCSF School of Medicine, Obstetrics, Gynecology and Reproductive Sciences, Division of Gynecologic Oncology, San Francisco, CA; [8]UC San Diego Moores Cancer Center, Division of Gynecologic Oncology, La Jolla, CA; [9]University of Arizona Cancer Center at Dignity Health St. Joseph's Hospital and Medical Center, Division of Gynecologic Oncology, Department of Obstetrics and Gynecology, Phoenix, AZ; [10]Memorial Sloan Kettering Cancer Center, Gynecologic Medical Oncology Service, New York, NY.

Presented at the American Society of Clinical Oncology, Chicago, IL, June 3-7, 2016

INTRODUCTION

- Cervical cancer is largely a human papillomavirus (HPV)-driven disease, the fourth most common cancer, and the most common cause of mortality in women worldwide[1]
- In the setting of recurrence outside of the central pelvis and non-surgically resectable persistence it is lethal, with a median overall survival (OS) of only 13–17 months among patients with access to first-line standard of care, platinum-based doublet chemotherapy +/– bevacizumab[1,2]
- There is no treatment that has demonstrated a survival benefit for patients with persistent/recurrent metastatic cervical cancer (PRmCC) whose disease has progressed after ≥1 lines of systemic-dose chemotherapy
 - Historical 12-month OS in trials evaluating this patient population has never exceeded 30% (**Figure 1**)[3-15]

Figure 1. Twelve-month survival rates in pretreated PRmCC the GOG experience.[3-15]

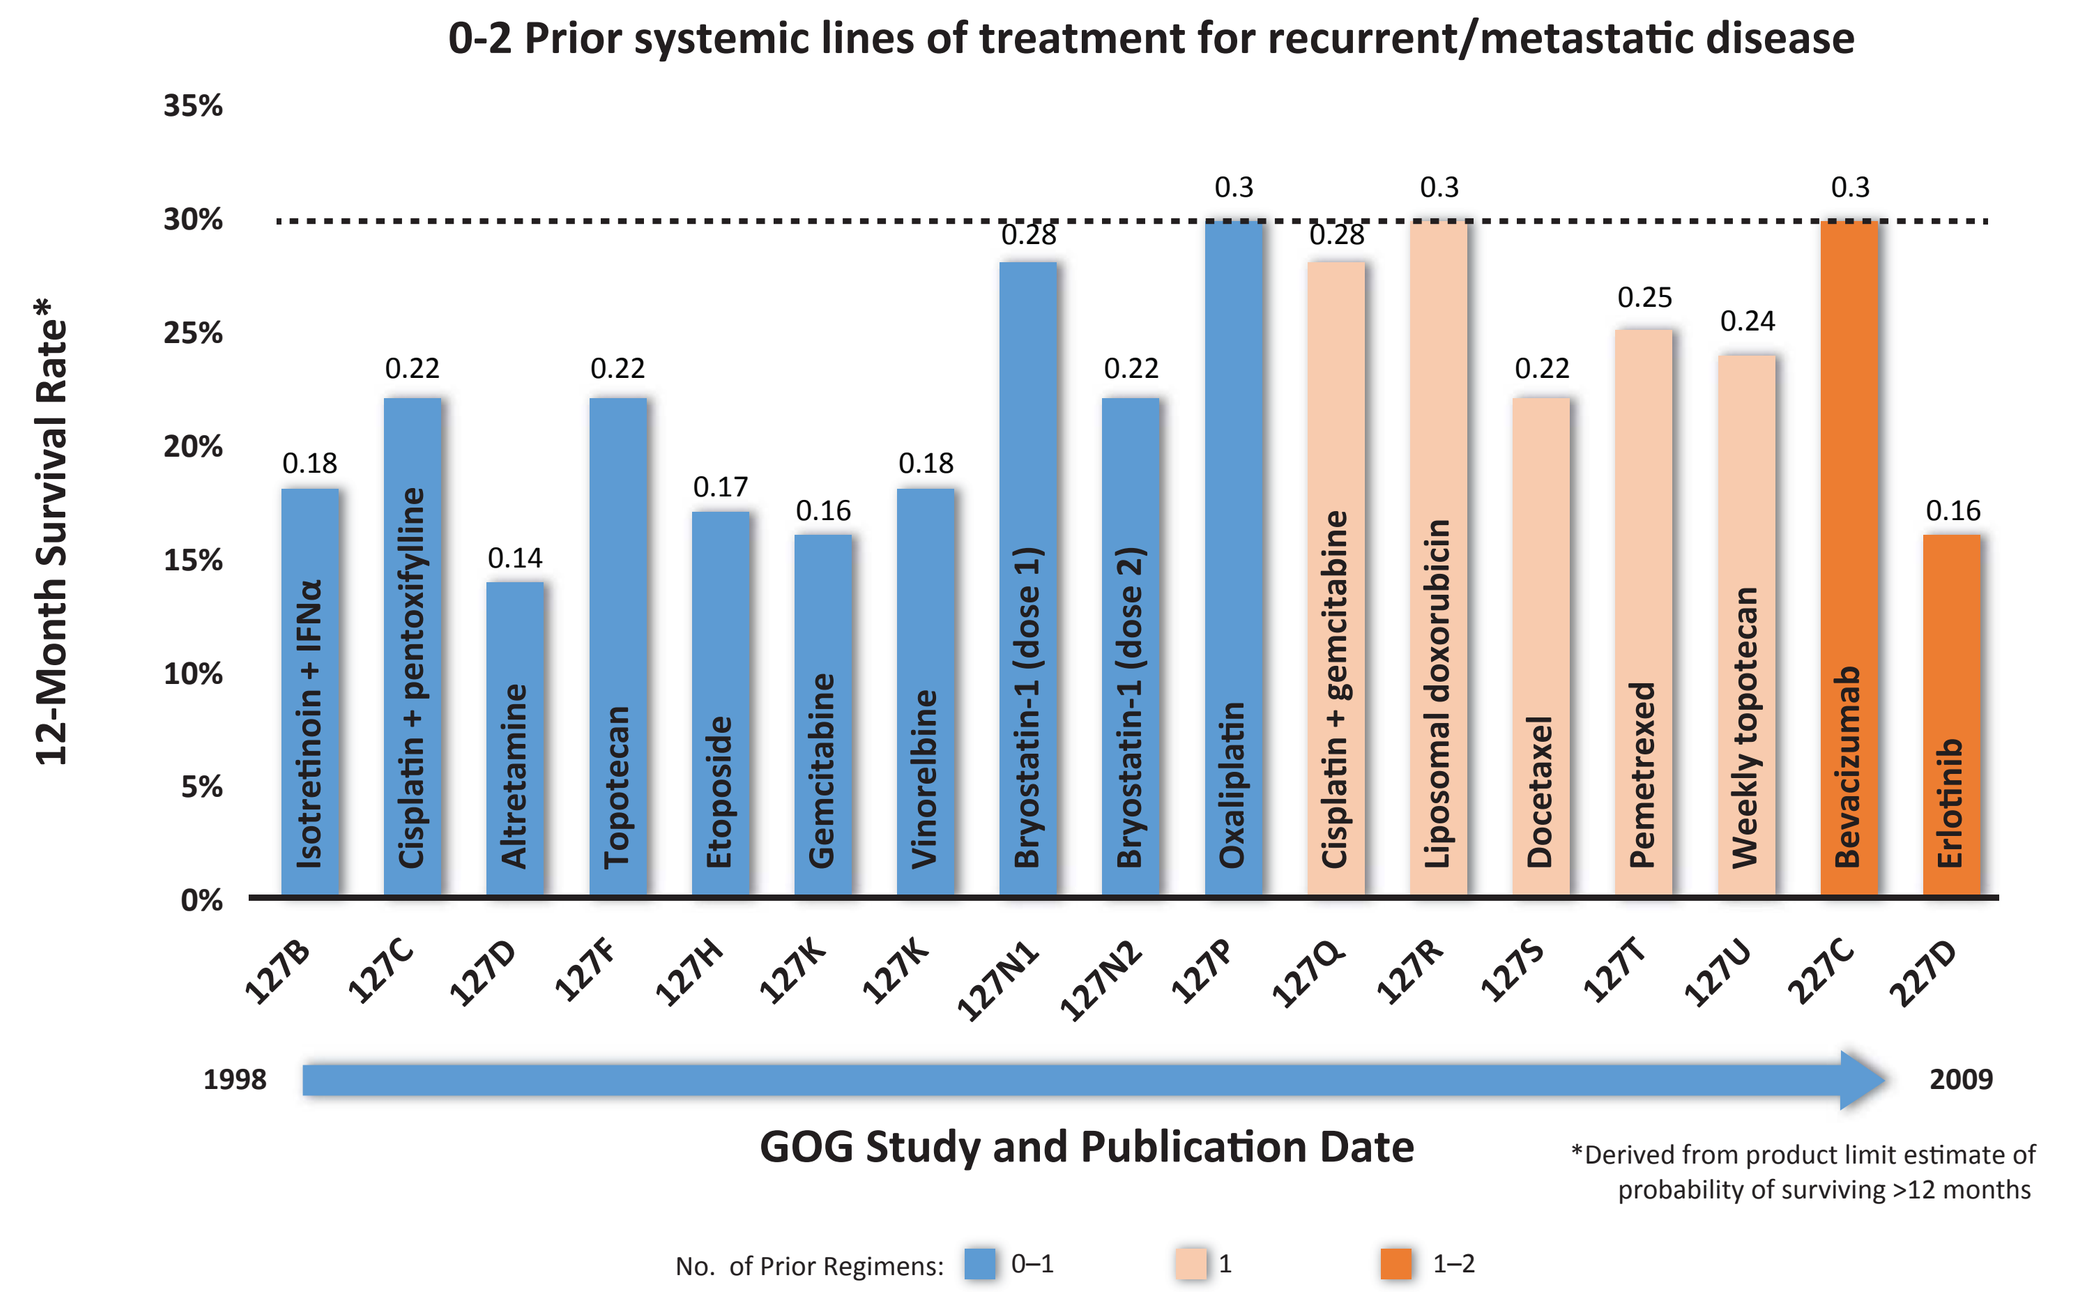
0-2 Prior systemic lines of treatment for recurrent/metastatic disease
12-Month Survival Rate*
35%
30%
25%
20%
15%
10%
5%
0%
Isotretinoin + IFNα 0.18
Cisplatin + pentoxifylline 0.22
Altretamine 0.14
Topotecan 0.22
Etoposide 0.17
Gemcitabine 0.16
Vinorelbine 0.18
Bryostatin-1 (dose 1) 0.28
Bryostatin-1 (dose 2) 0.22
Oxaliplatin 0.3
Cisplatin + gemcitabine 0.28
Liposomal doxorubicin 0.3
Docetaxel 0.22
Pemetrexed 0.25
Weekly topotecan 0.24
Bevacizumab 0.3
Erlotinib 0.16
127B 127C 127D 127F 127H 127K 127K 127N1 127N2 127P 127Q 127R 127S 127T 127U 227C 227D
1998
2009
GOG Study and Publication Date
*Derived from product limit estimate of probability of surviving >12 months
No. of Prior Regimens: 0–1 1 1–2

GOG, Gynecologic Oncology Group; IFNα, interferon alfa; PRmCC, persistent/recurrent metastatic cervical cancer.

RATIONALE FOR ADXS11-001 IN CERVICAL CANCER

- ADXS11-001 is a live, attenuated *Listeria monocytogenes* (Lm) immunotherapy bioengineered to secrete an HPV-E7 tumor antigen fused with a truncated fragment of listeriolysin O
 - This secreted HPV-E7 tumor antigen targets HPV-transformed cells (eg, cervical cancer), inducing antitumor T-cell immunity and breaking immune tolerance in the tumor microenvironment (**Figure 2**)[16]
- A randomized phase II trial of ADXS11-001 +/– cisplatin in patients with PRmCC and 0–2 prior lines of systemic therapy demonstrated promising activity (12-month OS = 32%) and acceptable toxicity[17]
- Herein we present stage 1 and preliminary stage 2 results from a phase II GOG/NRG trial of ADXS11-001 in patients with PRmCC who have received ≥1 prior lines of systemic-dose therapy

Figure 2. Step-by-step *Lm*-LLO immunomodulation.

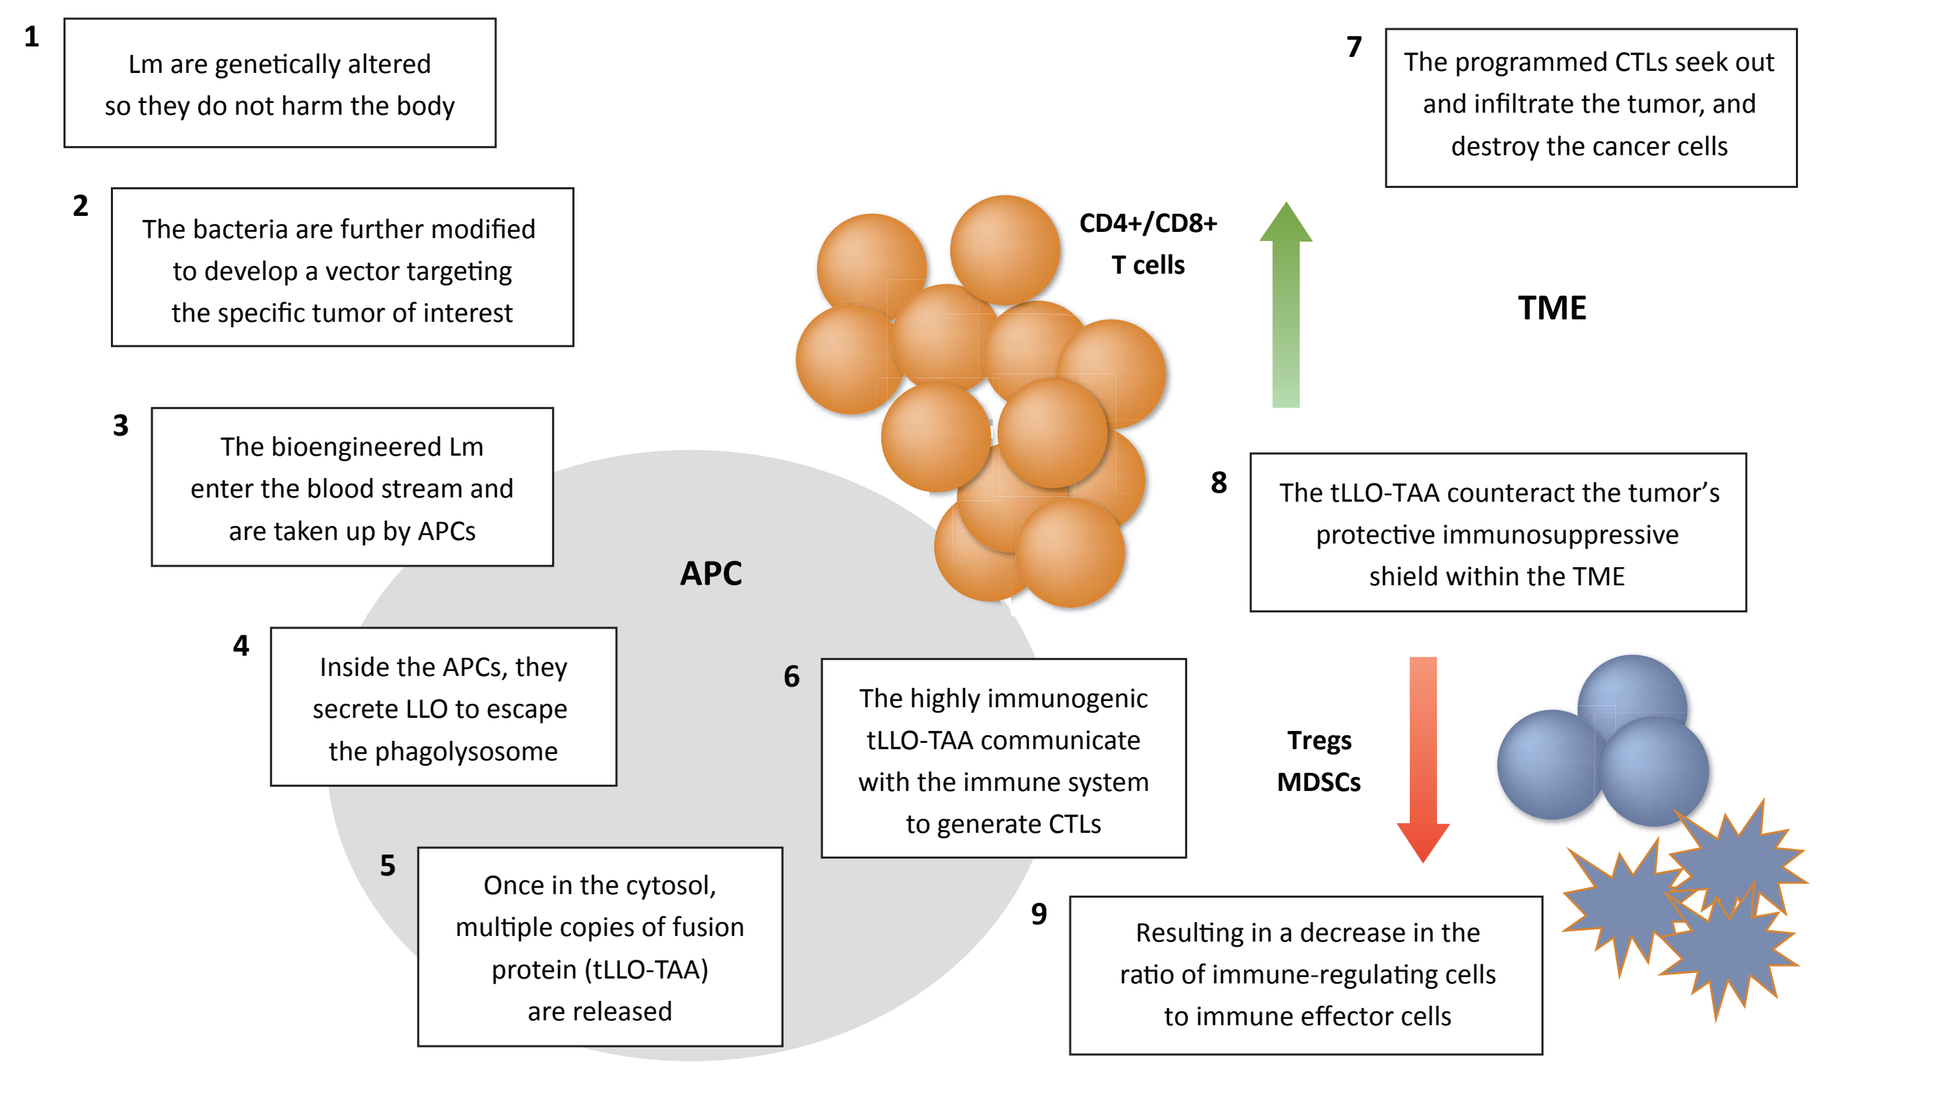
1 Lm are genetically altered so they do not harm the body
2 The bacteria are further modified to develop a vector targeting the specific tumor of interest
3 The bioengineered Lm enter the blood stream and are taken up by APCs
4 Inside the APCs, they secrete LLO to escape the phagolysosome
5 Once in the cytosol, multiple copies of fusion protein (tLLO-TAA) are released
6 The highly immunogenic tLLO-TAA communicate with the immune system to generate CTLs
7 The programmed CTLs seek out and infiltrate the tumor, and destroy the cancer cells
8 The tLLO-TAA counteract the tumor's protective immunosuppressive shield within the TME
9 Resulting in a decrease in the ratio of immune-regulating cells to immune effector cells
APC
CD4+/CD8+ T cells
TME
Tregs MDSCs

APCs, antigen-presenting cells; CD, cluster of differentiation; CTL, cytotoxic T lymphocyte; LLO, listeriolysin O; *Lm, Listeria monocytogenes*; MDSCs, myeloid-derived suppressor cells; TAA, tumor-associated antigen; tLLO, truncated LLO; TME, tumor microenvironment; Tregs, T-regulatory cell.

METHODS

STUDY DESIGN AND INTERVENTIONS

- GOG/NRG-0265 is a single-arm, 2-stage, phase II multicenter study (NCT01266460)
- ADXS11-001 at 1×10⁹ colony-forming units given on day 1 every 28 days is administered as a 250-mL infusion over 60 minutes
 - Patients initially received a maximum of 3 doses and were followed for clinical progression, confirmed radiologic disease progression, intolerable toxicity, or patient refusal of treatment
 - Following preliminary analysis of stage 1, the study was amended to allow for continuous (>3 doses) treatment with ADXS11-001 at 28-day intervals, until clinical progression, confirmed radiologic disease progression, intolerable toxicity, or patient refusal of treatment
- All patients receive antihistamine, anti-inflammatory, and antiemetic premedication, and a 7-day course of oral antibiotic therapy starting approximately 72 hours after each ADXS11-001 infusion
- The study schema, endpoints, and key eligibility criteria are depicted in **Figure 3**

Figure 3. GOG/NRG study schema and key eligibility requirements.

- N = ~63 Simon two-stage design
- ≥ 18 years
- Persistent/recurrent metastatic (PRmCC) squamous/non-squamous cervical cancer
- ≥ 1 prior line of systemic dose therapy for PRmCC, *excluding that received as a component ofprimary curative treatment*
- Prior bevacizumab allowed, but not required
- GOG PS 0/1
- Measurable disease ≥ 1 target lesion (RECIST 1.1)

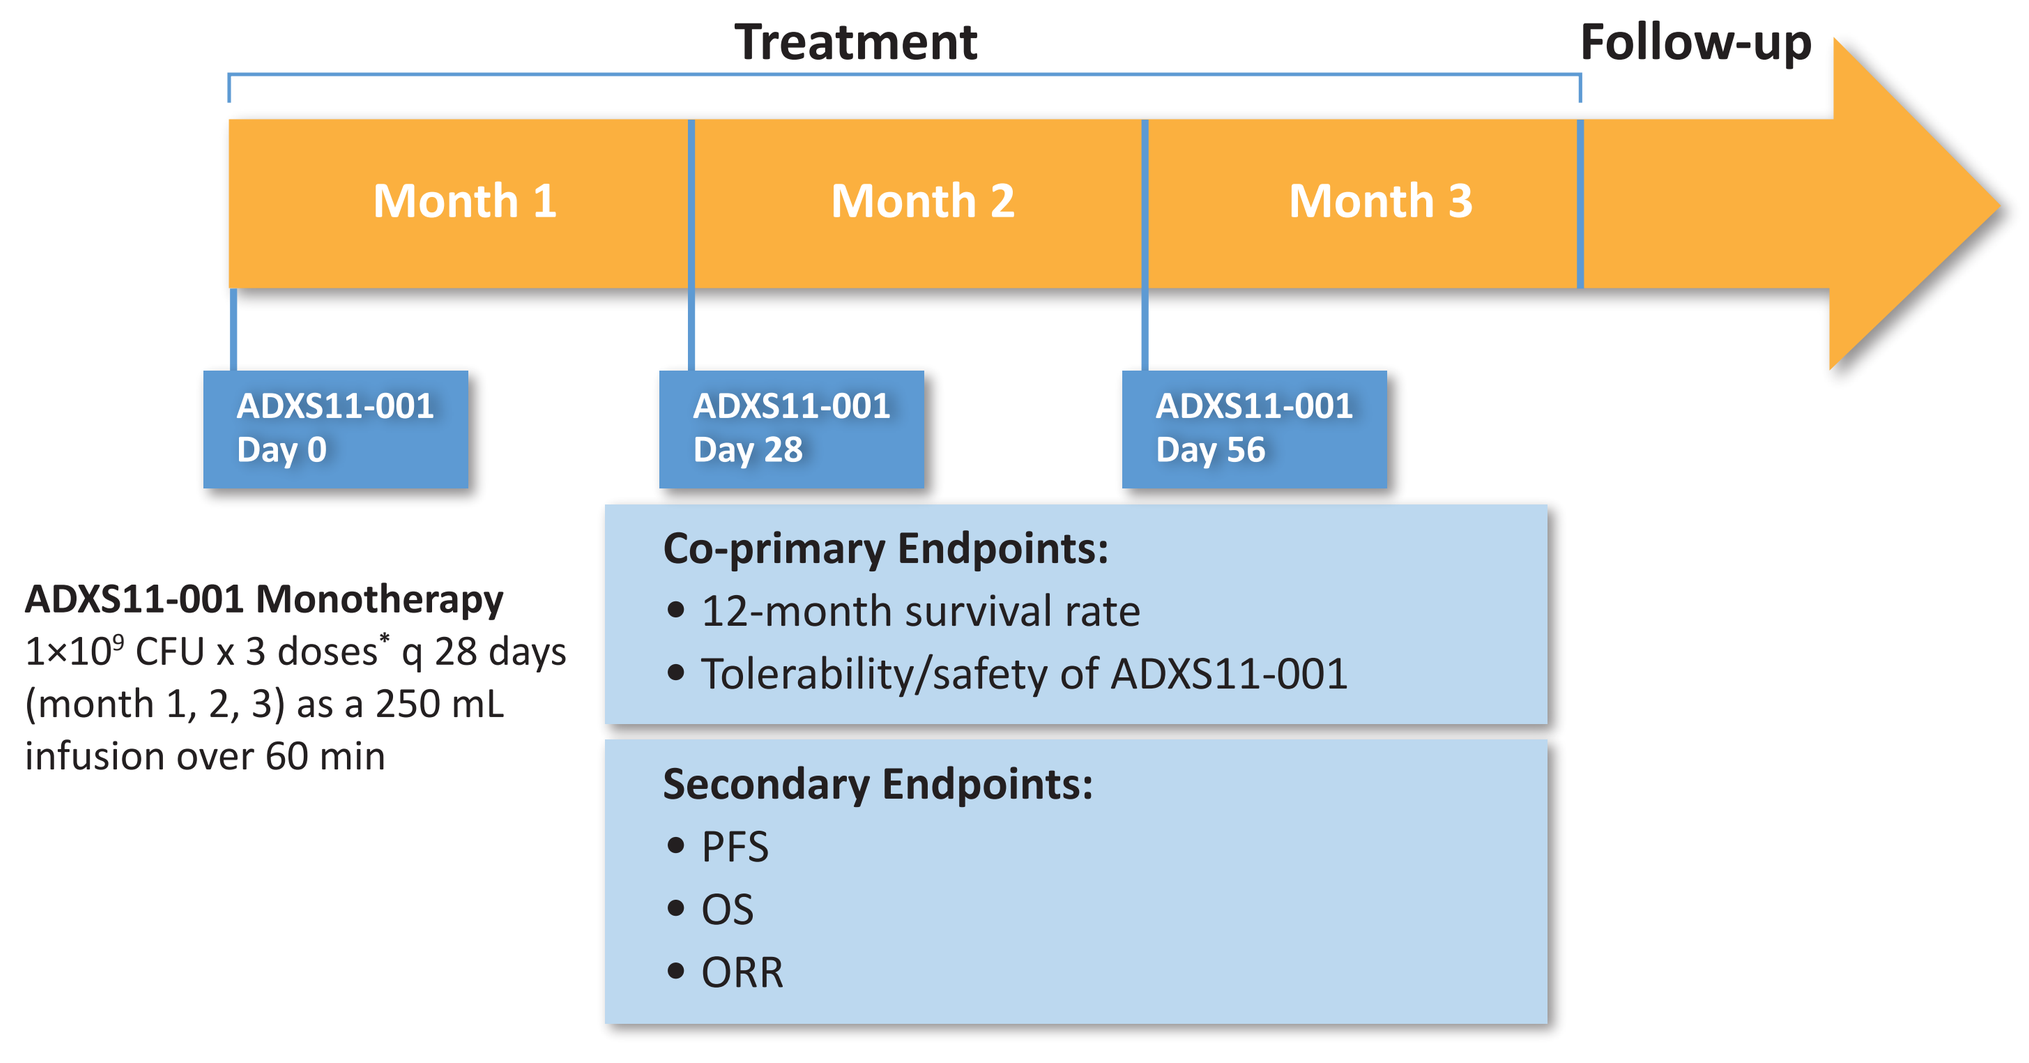
Treatment
Follow-up
Month 1
Month 2
Month 3
ADXS11-001 Day 0
ADXS11-001 Day 28
ADXS11-001 Day 56
ADXS11-001 Monotherapy
1×10⁹ CFU x 3 doses* q 28 days (month 1, 2, 3) as a 250 mL infusion over 60 min
Co-primary Endpoints:
• 12-month survival rate
• Tolerability/safety of ADXS11-001
Secondary Endpoints:
• PFS
• OS
• ORR

*Stage 2 amended to allow continuous (>3) dosing of ADXS11-001.

CFU, colony-forming units; GOG, Gynecologic Oncology Group; ORR, objective response rate; OS, overall survival; PFS, progression-free survival; PS, performance status; RECIST, Response Evaluation Criteria In Solid Tumors.

STATISTICAL METHODOLOGY

- Sample size calculation is based on the expected null proportion of patients surviving 12 months across historical trials of 20%
- Statistical tests with a 90% power are used to detect a 15% increase in 12-month survival (20% or 30%) at a one-sided significance level of 0.10
 - Targeted sample size is 27 patients (including 6 patients from safety lead-in) in stage 1 and 36 patients in stage 2
- The study was designed to proceed to stage 2 enrollment if conditional power at the end of stage 1 is ≥20%
- Stage 2 enrollment was successfully initiated on February 25, 2015
- Of note, all trials of ADXS11-001 were placed on clinical hold by the US Food and Drug Administration on October 6, 2015, for investigation of an isolated safety concern. The hold was subsequently lifted on December 15, 2015; however, enrollment to GOG/NRG-0265 did not resume at that time in favor of preliminary analysis of the stage 2 cohort to inform further study of ADXS11-001 in PRmCC

RESULTS

PATIENT POPULATION AND TREATMENT EXPOSURE

- The CONSORT diagram (**Figure 4**) depicts the total number of patients enrolled and subsequently treated in stages 1 and 2, as well as distribution of ADXS11-001 doses received
 - Of the 26 treated patients in stage 1, median number of doses received was 3 (range 1–3), and 69% (n = 18) received the maximum 3 doses
 - Of the 24 treated patients in stage 2, the median number of doses received was 2.5 (range 1–6), and 50% (n = 12) received ≥3 doses
 - At the time of the clinical hold, 10 patients were actively receiving ADXS11-001
 - Of these, 4 (40%) patients received ≥3 doses, while 6 (60%) patients received <3 doses

The baseline demographics and clinical characteristics of patients enrolled in stage 1 and in stage 2 are presented in **Table 1**

Figure 4. CONSORT diagram.

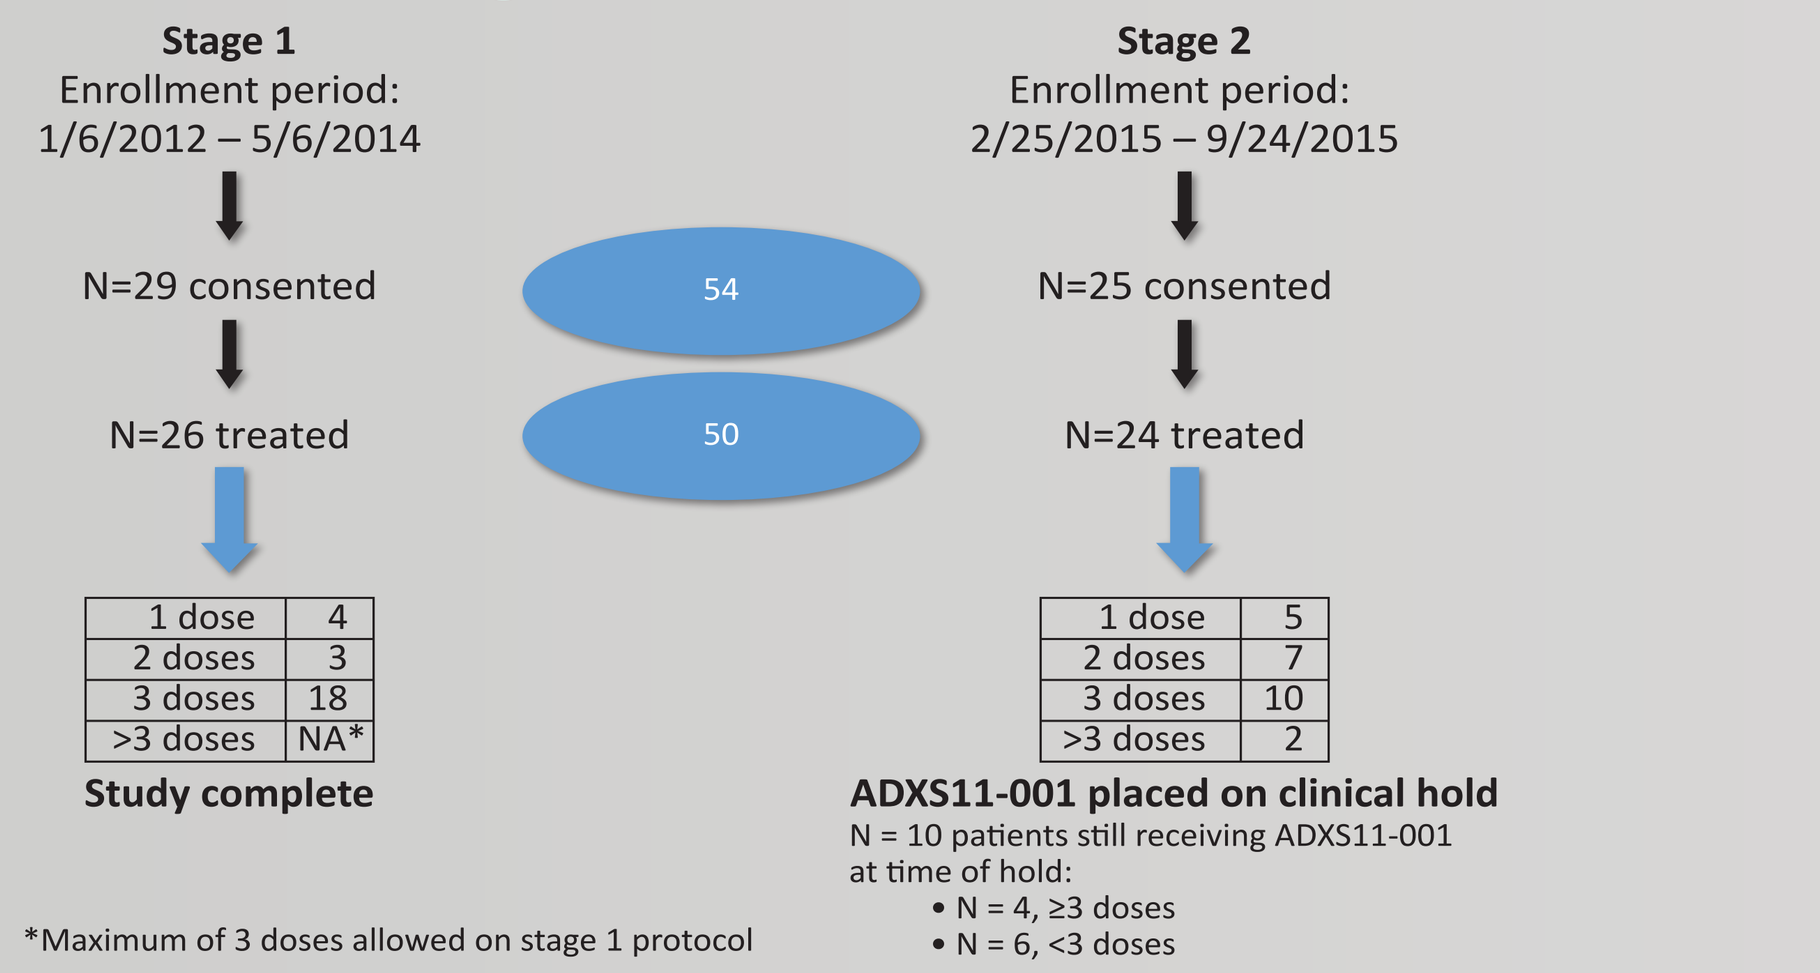
Stage 1
Enrollment period: 1/6/2012 – 5/6/2014
N=29 consented
N=26 treated
1 dose 4
2 doses 3
3 doses 18
>3 doses NA*
Study complete
54
50
Stage 2
Enrollment period: 2/25/2015 – 9/24/2015
N=25 consented
N=24 treated
1 dose 5
2 doses 7
3 doses 10
>3 doses 2
ADXS11-001 placed on clinical hold
N = 10 patients still receiving ADXS11-001 at time of hold:
• N = 4, ≥3 doses
• N = 6, <3 doses
*Maximum of 3 doses allowed on stage 1 protocol

Table 1. Baseline demographics and clinical characteristics

	Stage 1 (n = 26)	Stage 2 (n = 24)
Median age (range), years	46.5 (33–66)	42 (29–70)
Race, n (%) White	18 (69)	19 (79)
GOG PS, n (%) 0 vs 1	16 (61.5) vs 10 (38.5)	15 (62.5) vs 9 (37.5)
FIGO stage at diagnosis, n (%)		
IA	0	1 (4)
IB	11 (42)	7 (29)
IIA	3 (11)	0
IIB	6 (23)	8 (33)
IIIB	2 (8)	2 (8)
IVN	4 (16)	6 (25)
NA	0 (0)	1 (4)
Prior lines of systemic-dose therapy, n (%)		
1	10 (38)	14 (58)
2	12 (46)	10 (42)
3	4 (16)	0 (0)
Prior bevacizumab, n (%)	8 (31)	20 (83)
Prior pelvic radiation, n (%)	21 (81)	22 (92)

FIGO, Fédération Internationale de Gynécologie et d'Obstétrique; GOG, Gynecologic Oncology Group; PS, performance status.

EFFICACY

- For the patients treated in stage 1 (n = 26) of the study
 - The 12-month survival rate is 38.5% (n = 10/26)
 - Patients alive at 12 months (n = 10/26) had received 1 (3/8 patients), 2 (6/14 patients), or 3 (1/4 patients) prior lines of systemic-dose therapy, indicating that the 12-month survival rate was achieved irrespective of extent of prior therapy
 - Median OS is 7.7 months (95% confidence interval [CI]: 3.9–12.4) and median progression-free survival (PFS) is 3.1 months (95% CI: 2.8–3.7; **Figure 5A**)
 - Investigator assessment of tumor best response was reported in 20/26 treated patients
 - Seven patients (27%) experienced stable disease (SD) and 10 patients (38%) had progressive disease (PD)
 - The remaining 6 patients were not evaluable for response per investigator
 - Post-hoc exploratory analysis of the 18/26 patients (69%) who received all 3 per-protocol doses of ADXS11-001 shows a median OS >1 year (12.1 [95% CI: 6.8–not reached (NR)] months), and 12-month survival rate of 55.6% (**Figure 5B**)
 - Twelve-month survival in GOG/NRG-0265 compares favorably to the historical GOG clinical trials series in PRmCC[3-15,18] (**Figure 5C**)

Figure 5. Survival of patients treated in stage 1 of the study: A) 12-month survival rate, OS, and PFS; B) exploratory analysis of OS in patients receiving all 3 of the per-protocol doses of ADXS11-001; and C) GOG/NRG-0265 survival in the context of historical GOG PRmCC clinical trials.

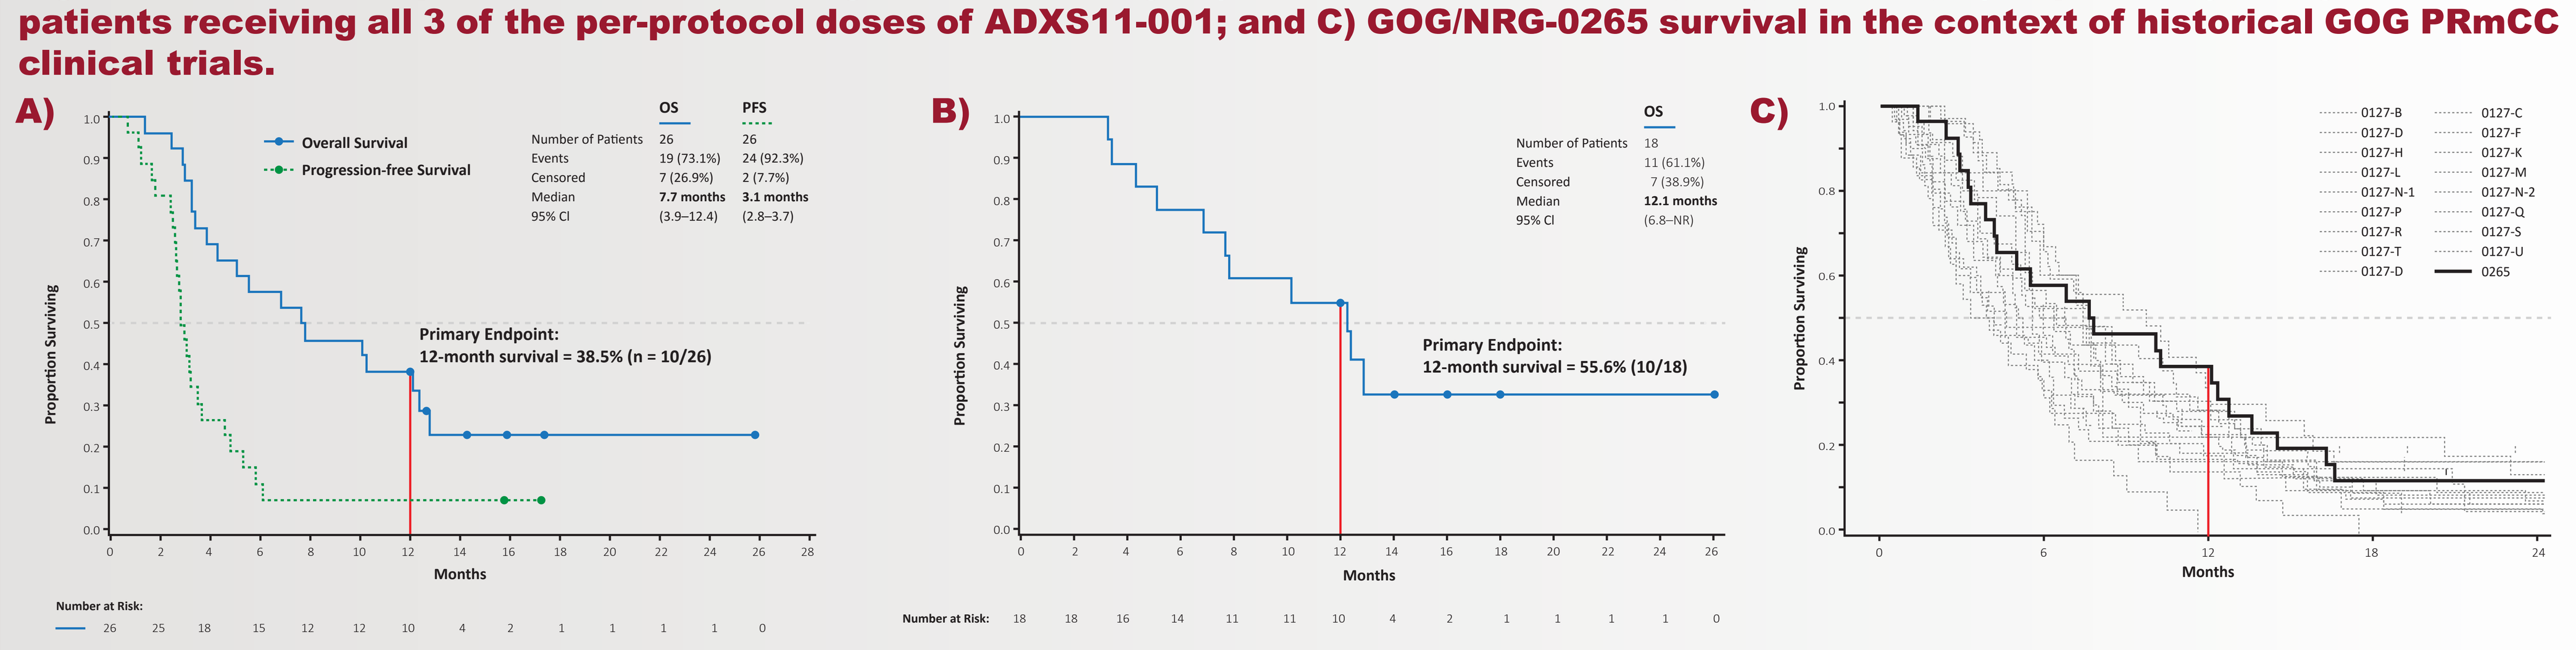
A)
Overall Survival
Progression-free Survival
OS PFS
Number of Patients 26 26
Events 19 (73.1%) 24 (92.3%)
Censored 7 (26.9%) 2 (7.7%)
Median 7.7 months 3.1 months
95% CI (3.9–12.4) (2.8–3.7)
Primary Endpoint:
12-month survival = 38.5% (n = 10/26)
Proportion Surviving
Months
Number at Risk:
26 25 18 15 12 12 10 4 2 1 1 1 1 0
26 22 7 3 2 2 2 2 1 0
B)
OS
Number of Patients 18
Events 11 (61.1%)
Censored 7 (38.9%)
Median 12.1 months
95% CI (6.8–NR)
Primary Endpoint:
12-month survival = 55.6% (10/18)
Proportion Surviving
Months
Number at Risk: 18 18 16 14 11 11 10 4 2 1 1 1 1 0
CI, confidence interval; OS, overall survival; PFS, progression-free survival.
C)
0127-B 0127-C 0127-D 0127-F 0127-H 0127-K 0127-L 0127-M 0127-N-1 0127-N-2 0127-P 0127-Q 0127-R 0127-S 0127-T 0127-U 0127-D 0265
Proportion Surviving
Months

CI, confidence interval; GOG, Gynecologic Oncology Group; OS, overall survival; PFS, progression-free survival; PRmCC, persistent/recurrent metastatic cervical cancer; NR, not reached.

SAFETY/TOLERABILITY

- All patients (26; 100%) treated in stage 1 of the study had at least 1 adverse event (AE)
 - In 24 (92%) patients these AEs were treatment related (TRAEs): 19 (73%) had grade 1–2 TRAEs only, 4 (15%) had grade 3 TRAEs, and 1 (4%) patient had a possibly related grade 4 TRAE
 - The most common TRAEs >30% were fatigue, chills, fever, nausea, and headache (**Table 2**)
- Safety findings among patients enrolled in stage 2 are similar to those reported in detail for stage 1, and are not shown here

Table 2. TRAEs recorded in patients treated in stage 1 of the study

Adverse Event Summary (n = 26)			
AE	Grade 1–4	Grade 3	Grade 4
Patients with ≥1 TRAE, n (%)	24 (92)	4 (15)	1 (4)*
TRAEs occurring in ≥10% of patients			
Fatigue	15 (58)	-	-
Chills	14 (54)	-	-
Fever	11 (42)	-	-
Nausea	10 (39)	-	-
Headache	9 (35)	-	-
Hypotension	7 (27)	2 (8)	-
Vomiting	6 (23)	-	-
Cytokine release syndrome	5 (19)	3 (12)	-
Myalgia	5 (19)	-	-
Abdominal pain	4 (15)	-	-
General pain	4 (15)	-	-
Flu-like symptoms	3 (11)	-	-
AST elevation	3 (11)	-	-

*The observed grade 4 TRAE recorded in 1 patient (lung infection and sepsis) was considered *possibly related* to treatment. AE, adverse event; AST, aspartate aminotransferase; TRAE, treatment-related adverse event.

- For the patients treated in stage 2 (n = 24) of the study
 - The primary endpoint 12-month survival rate cannot be calculated due to limited median follow-up of 8.7 months
 - The 6-month survival rate is 42% (10/24) and median OS is 4.8 months (95% CI: 3.6–NR) (Figure 6A)
 - Although 10/24 (42%) patients discontinued ADXS11-001 without progression or death due to clinical hold, median PFS is 2.6 months (95% CI: 2.0–3.2) (**Figure 6A**), similar to that observed in stage 1
 - Among the 50% of patients (12/24) who received 3 or more doses of ADXS11-001, median OS is NR (95% CI: 3.5–NR) with median follow-up of 9.2 months, and 6-month survival rate is 67% (**Figure 6B**)
 - Investigator assessment of tumor best response was reported in 20/24 treated patients
 - One (4%) patient experienced a complete response (CR), 8 (33%) experienced SD, and 11 (46%) had PD
 - Clinical history and images representative of the durable CR are shown in **Figure 7**

Figure 6. Survival of patients treated in stage 2 of the study: A) 6-month survival rate, OS, and PFS; and B) exploratory analysis of OS in patients receiving ≥3 doses of ADXS11-001.

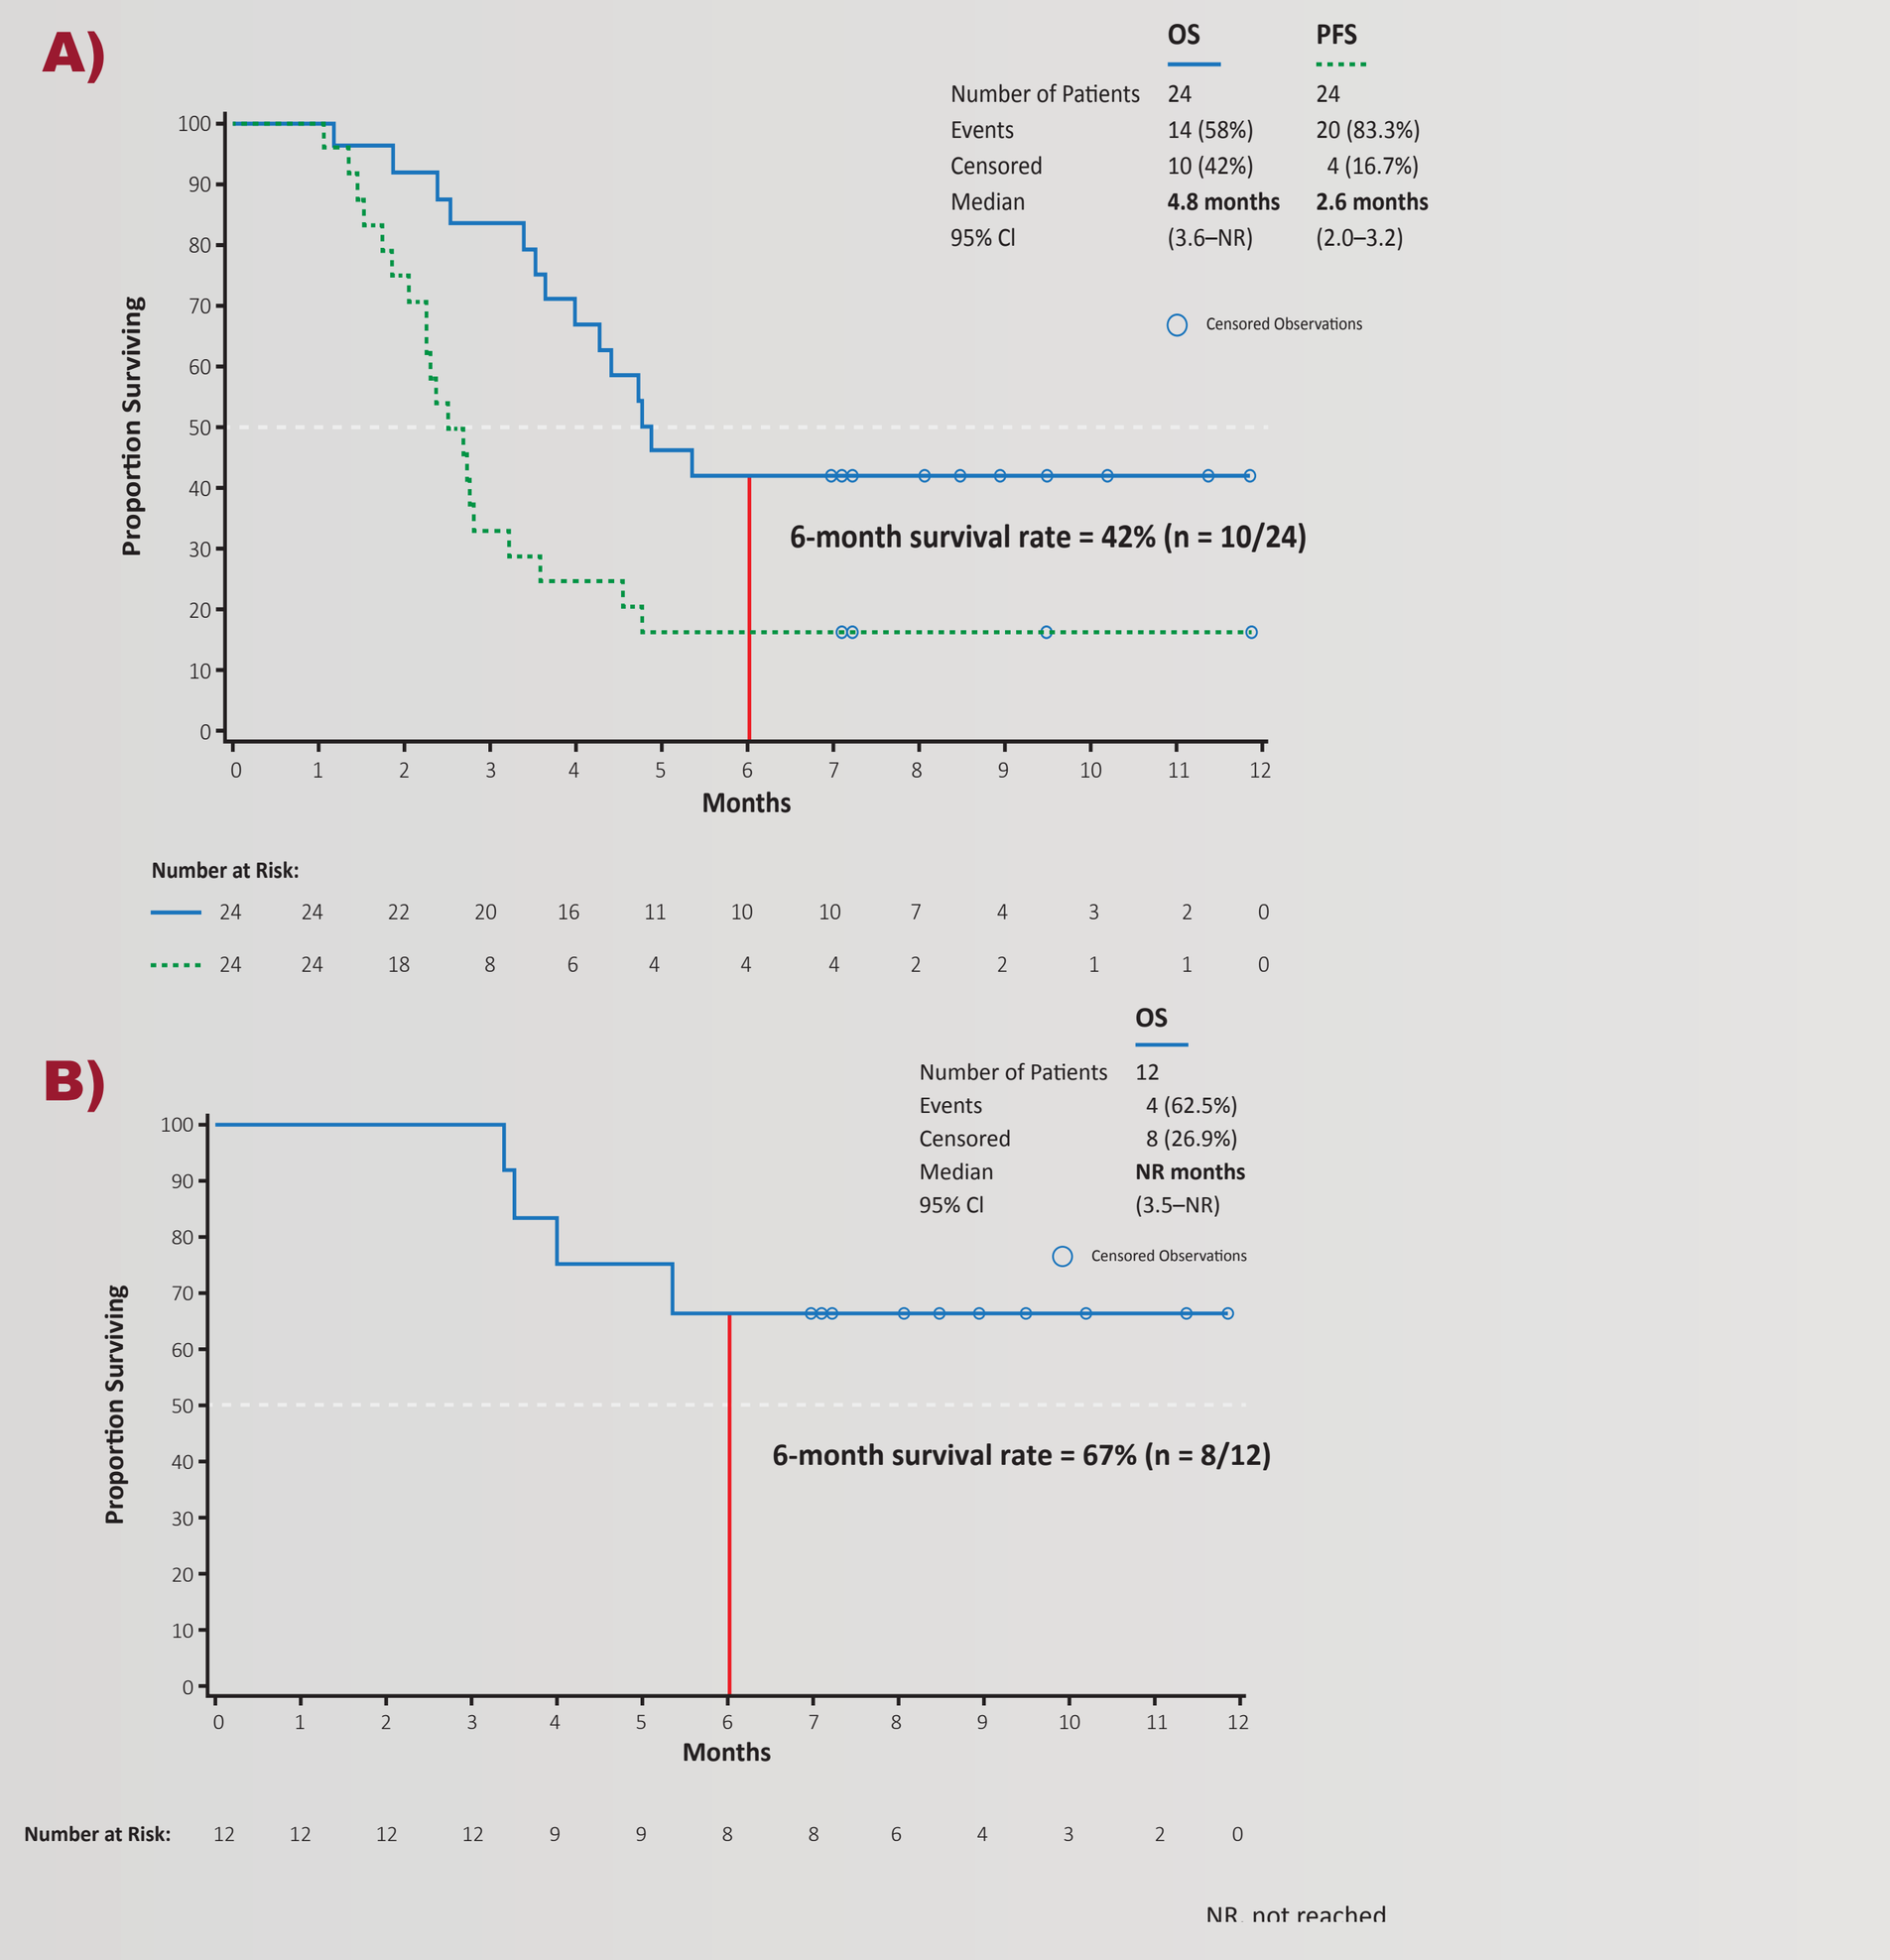
A)
OS PFS
Number of Patients 24 24
Events 14 (58%) 20 (83.3%)
Censored 10 (42%) 4 (16.7%)
Median 4.8 months 2.6 months
95% CI (3.6–NR) (2.0–3.2)
Censored Observations
6-month survival rate = 42% (n = 10/24)
Proportion Surviving
Months
Number at Risk:
24 24 22 20 16 11 10 10 7 4 3 2 0
24 24 18 8 6 4 4 4 2 2 1 1 0
B)
OS
Number of Patients 12
Events 4 (62.5%)
Censored 8 (26.9%)
Median NR months
95% CI (3.5–NR)
Censored Observations
6-month survival rate = 67% (n = 8/12)
Proportion Surviving
Months
Number at Risk: 12 12 12 12 9 9 8 8 6 4 3 2 0
NR, not reached

CI, confidence interval; OS, overall survival; NR, not reached; PFS, progression-free survival.

Figure 7. GOG/NRG-0265 stage 2 case study – durable CR to ADXS11-001.

- 66-year-old woman diagnosed with squamous cell cancer of the cervix in 2006, surgically treated with radical hysterectomy in 2007
- Pelvic recurrence in 2014
 - Paclitaxel/carboplatin × 8 cycles (6 cycles with bevacizumab) → cisplatin (2 cycles) + pelvic radiation. Treatment completed August 2014
- Systemic recurrence June 2015
 - Enrolled in GOG/NRG-0265

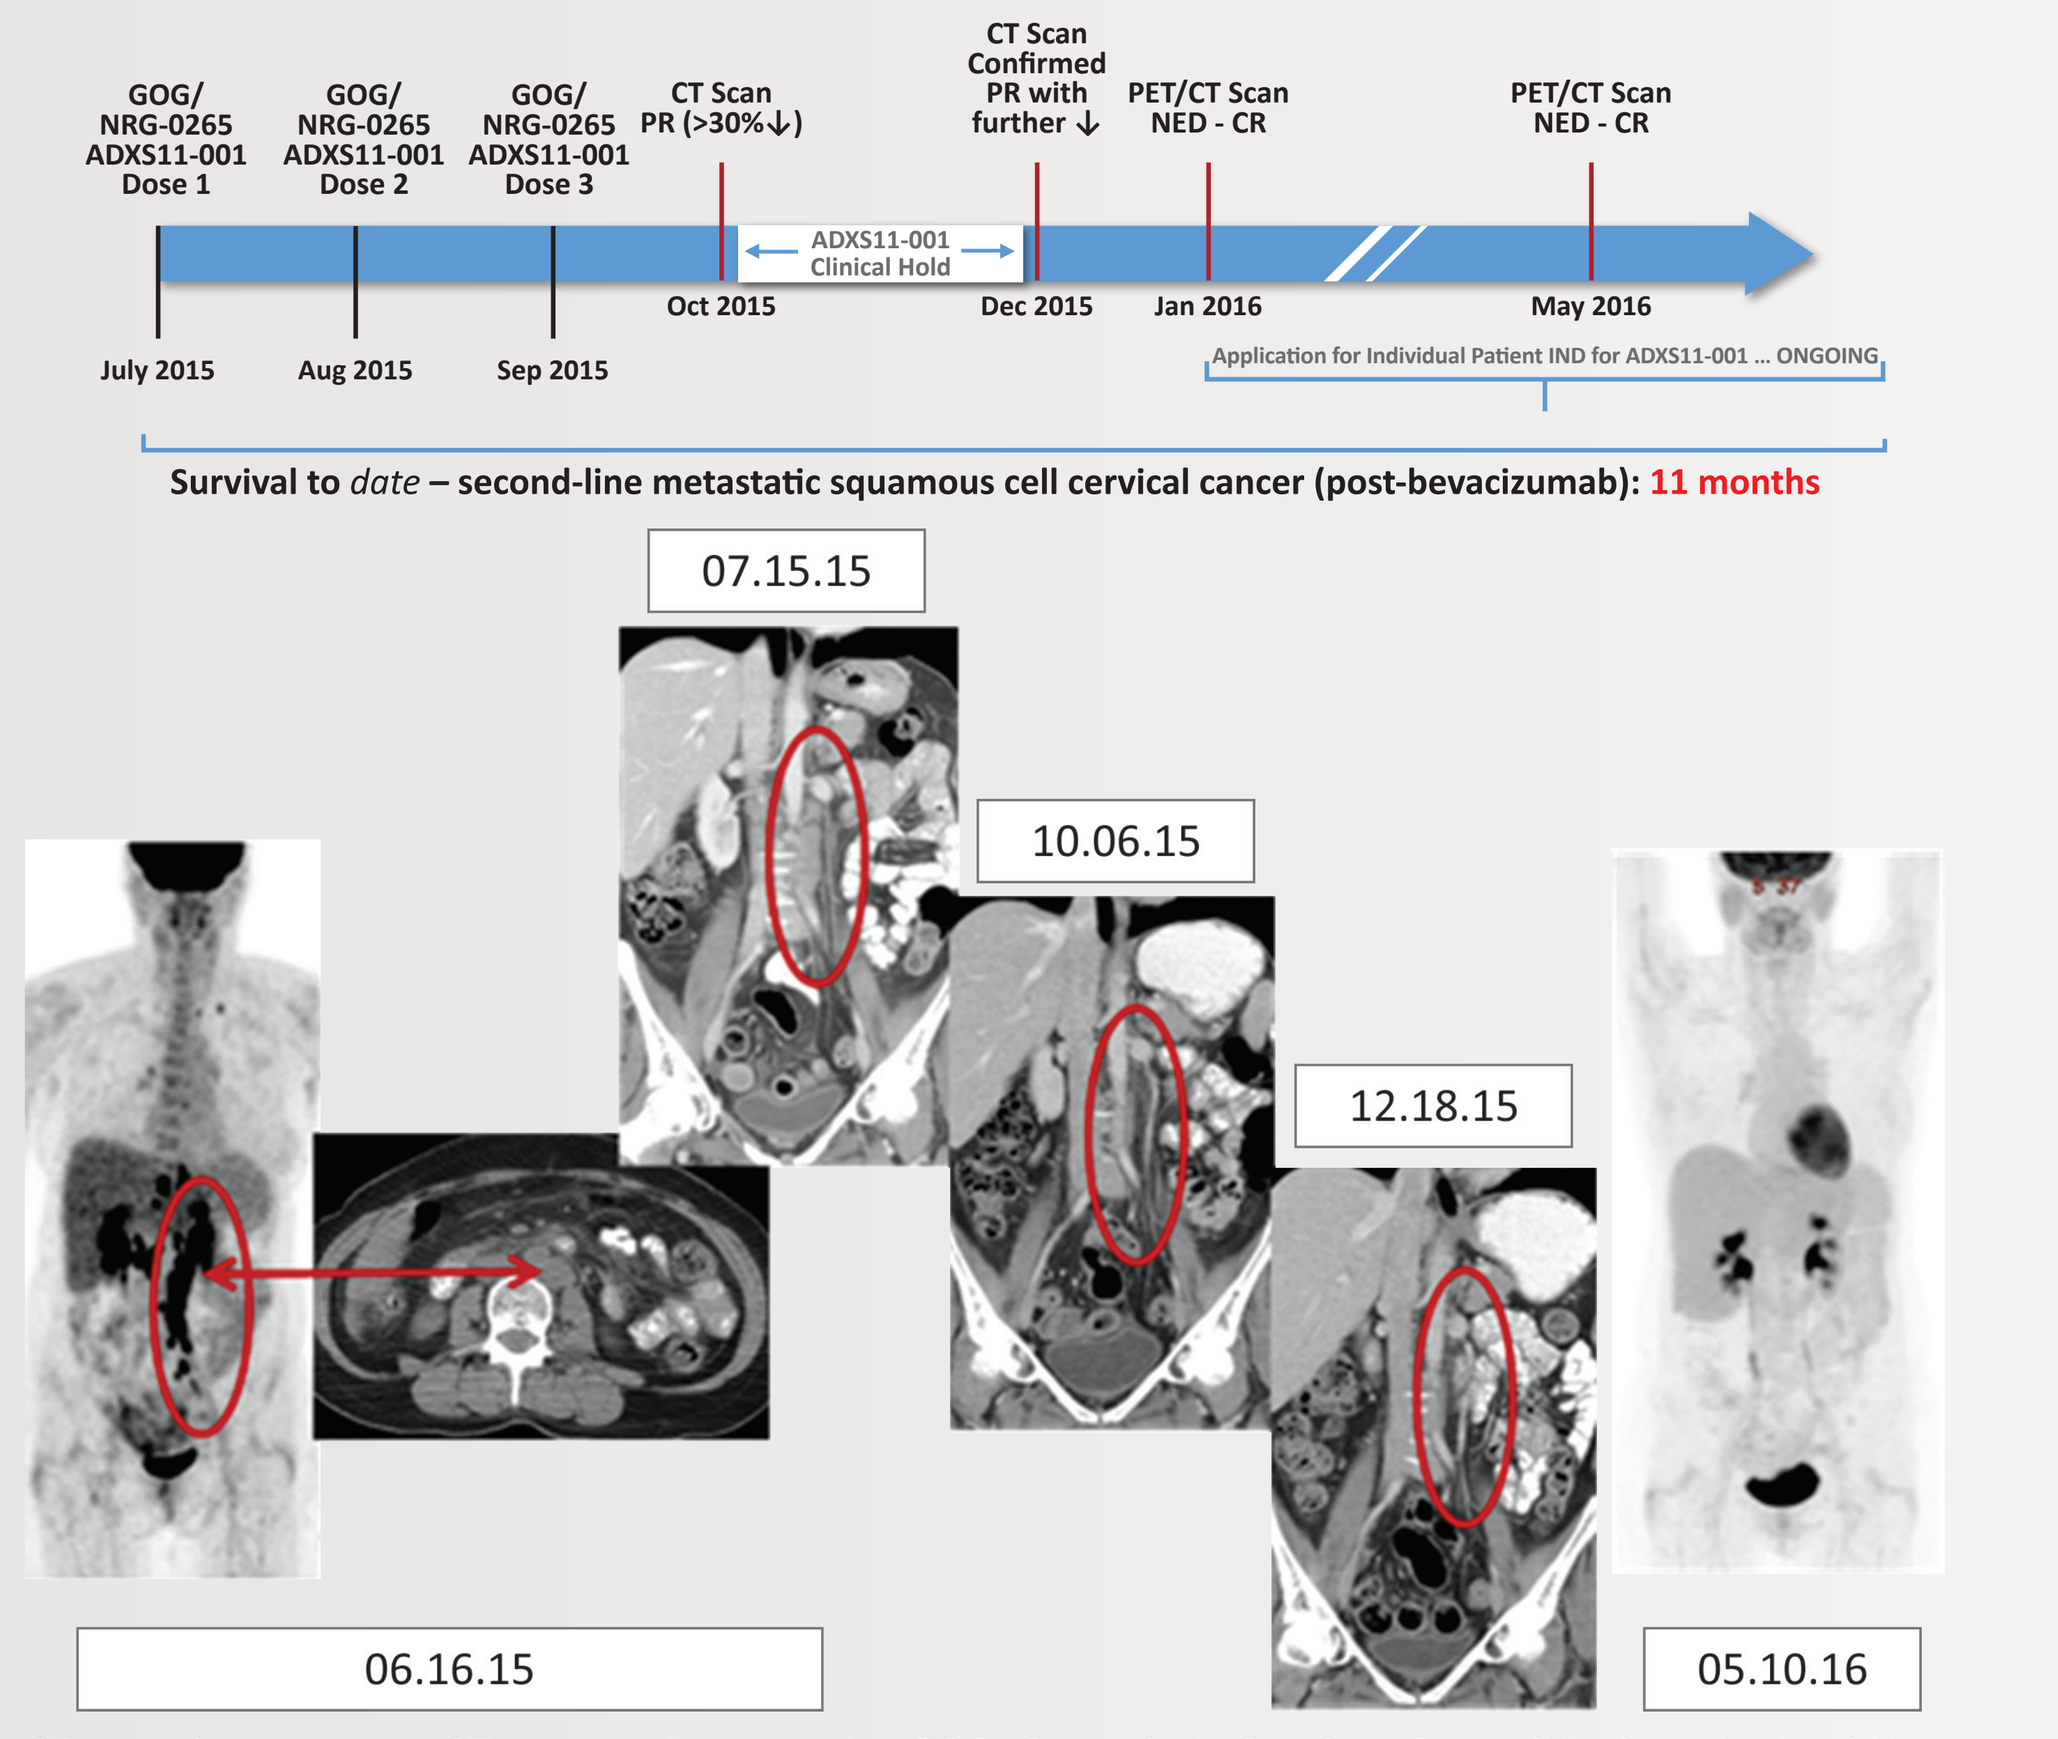
GOG/NRG-0265 ADXS11-001 Dose 1
GOG/NRG-0265 ADXS11-001 Dose 2
GOG/NRG-0265 ADXS11-001 Dose 3
CT Scan PR (>30%↓)
ADXS11-001 Clinical Hold
CT Scan Confirmed PR with further ↓
PET/CT Scan NED - CR
PET/CT Scan NED - CR
July 2015
Aug 2015
Sep 2015
Oct 2015
Dec 2015
Jan 2016
May 2016
Application for Individual Patient IND for ADXS11-001 ... ONGOING
Survival to date – second-line metastatic squamous cell cervical cancer (post-bevacizumab): 11 months
07.15.15
10.06.15
12.18.15
06.16.15
05.10.16

CR, complete response; CT, computed tomography; GOG, Gynecologic Oncology Group; IND, Investigational New Drug; NED, no evidence of disease; PET, positron emission tomography; PR, partial response.

CONCLUSION

- In patients with PRmCC and progression following ≥1 prior line of systemic therapy, ADXS11-001 is well tolerated and demonstrates a 38.5% rate of 12-month survival (n = 10/26)
- Although preliminary, findings from stage 2 reinforce the rationale for further controlled investigation of ADXS11-001 in PRmCC, and suggest consistent survival benefit in a heavily bevacizumab-pretreated population (31% vs 83% in stage 1 and stage 2, respectively), particularly among those patients receiving 3 or more doses of immunotherapy
- An international Advaxis-sponsored phase III study of ADXS11-001 as adjuvant treatment of high-risk locally advanced cervical cancer (AIM2CERV) is under development in collaboration with the GOG Foundation

REFERENCES

1. Ferlay J, et al. *Int J Cancer*. 2015;136(5):E359-E386.
2. Tewari KS, et al. *N Engl J Med*. 2014;370(8):734-743.
3. Tewari KS, Monk BJ. *Curr Oncol Rep*. 2005;7(6):419-434.
4. Muggia FM, et al. *Gynecol Oncol*. 2004;92(2):639-643.
5. Plaxe SC, et al. *Cancer Chemother Pharmacol*. 2002;50(2):151-154.
6. Armstrong DK, et al. *Invest New Drugs*. 2003;21(4):453-457.
7. Fracasso PM, et al. *Gynecol Oncol*. 2003;90(1):177-180.
8. Brewer CA, et al. *Gynecol Oncol*. 2006;100(2):385-388.
9. Rose PG, et al. *Gynecol Oncol*. 2006;102(2):210-213.
10. Garcia AA, et al. *Am J Clin Oncol*. 2007;30(4):428-431.
11. Miller DS, et al. *Gynecol Oncol*. 2008;110(1):65-70.
12. Fiorica JV, et al. *Gynecol Oncol*. 2009;115(2):285-289.
13. Monk BJ, et al. *J Clin Oncol*. 2009;27(7):1069-1074.
14. Schilder RJ, et al. *Int J Gynecol Cancer*. 2009;19(5):929-933.
15. National Cancer Institute. Vaccine therapy in treating patients with persistent or recurrent cervical cancer. http://www.cancer.gov/about-cancer/treatment/clinical-trials/search/view?cdrid=691288. Accessed May 25, 2016.
16. Krupar R, et al. Presented at the 107th Annual Meeting of the American Association for Cancer Research; April 16–20, 2016; New Orleans, LA. Abstract LB-095.
17. Basu P, et al. *J Clin Oncol*. 2014;32(suppl): abstract 5610.
18. Gynecologic Oncology Group/NRG Oncology. Data on file, 2016.

ACKNOWLEDGMENTS

Editorial and medical writing assistance was provided by Mary Smith, PhD CMPP, TRM Oncology, funded by Advaxis. The authors are fully responsible for all content and editorial decisions for this poster.

DISCLOSURES

W. Huh: Consulting or advisory role – TheVax, Invio (DSMB).
D. Dizon: Disclosure accurate in ASCO COI.
M.A. Powell: No conflicts of interest to disclose.
C.A. Leath III: Research funding – Novartis, AstraZeneca, Plexxikon, and Celsion. Supported in part by NIH 3P30CA013148-43S3.
L.M. Landrum: No conflicts of interest to disclose.
E. Tanner: No conflicts of interest to disclose.
R. Higgins: No conflicts of interest to disclose.
S. Ueda: No conflicts of interest to disclose.
M. McHale: Consulting or advisory role – Ethicon, for new homeostatic product. Research funding – Navidea, funded clinical trial, with no principal investigator compensation. Travel, accommodation, expenses – Ethicon.
B.J. Monk: Consulting or advisory role – Advaxis.
C. Aghajanian: No conflicts of interest to disclose.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

http://bit.ly/ASCOposter