P163

Phase 1/2 study of ADXS31-142 alone and in combination with pembrolizumab in patients with previously treated metastatic castration-resistant prostate cancer (mCRPC): The KEYNOTE-046 trial

N. Haas[1], M. Stein[2], R. Tutrone[3], R. Perini[4], A. Denker[4], D. Mauro[5], L. Fong[6]

[1]Abramson Cancer Center, University of Pennsylvania, Philadelphia, PA, USA; [2]Rutgers Cancer Institute of New Jersey, New Brunswick, NJ, USA; [3]Chesapeake Urology Research Associates, Towson, MD, USA; [4]Merck & Co., Inc., Kenilworth, NJ, USA; [5]Advaxis Inc., Princeton, NJ, USA; [6]University of California San Francisco, San Francisco, CA, USA

INTRODUCTION

- In 2015, the National Cancer Institute's Surveillance, Epidemiology, and End Results Program estimates there will be 220,800 new cases of PC and 27,540 deaths from the disease.[1]
 - Approximately 14% of men will be diagnosed with PC at some point during their lifetime[1]
- In recent years, treatment options for men with mCRPC have improved. However, the outlook for patients remains poor, with overall survival reported to be approximately 2 years.[2]
- PC generates prostate tumor-specific antigens such as prostate-specific antigen (PSA), prostatic acid phosphatase (PAP), and prostate-specific membrane antigen.[3]
 - Given that the prostate is a dispensable organ, any potential autoimmunity to prostate cells generated from immunotherapy does not impact patient safety, making directed immunotherapy a viable treatment option
 - Studies of immunotherapy in the treatment of PC have demonstrated improved survival (4.1-month improvement in median overall survival when compared to placebo)[4]
- ADXS31-142 is a live, attenuated, bioengineered *Listeria monocytogenes (Lm)*-listeriolysin O (LLO) immunotherapy designed to target PSA for the treatment of PC.[5-8]
- ADXS31-142 secretes an antigen-adjuvant fusion protein (tLLO-PSA) consisting of a truncated fragment of LLO (tLLO) fused to the human PSA, which is rapidly taken up by APCs, stimulating adaptive immunity and resulting in the generation of a new population of tumor antigen-specific cytotoxic T lymphocytes (CTLs).[9]
 - *Lm*-LLO immunotherapies stimulate both innate and adaptive tumor-specific immunity. They direct antigen-presenting cells (APCs) to stimulate and activate the immune system and also reduce tumor immunosuppression in the tumor microenvironment by neutralizing regulatory T cells (Tregs) and myeloid-derived suppressor cells (MDSCs) **(Figure 1)**
- Pembrolizumab (MK-3475), a humanized monoclonal antibody of the IgG4κ subclass, blocks the binding of programmed cell death protein-1 (PD-1) receptor to its ligands PD-L1 and PD-L2.[10]
- By blocking PD-1, pembrolizumab activates antigen-specific T cells including CD8-positive T cells (inhibition mediated via binding of PD-L1) as well as immune T cells in lymphoid organs (inhibition mediated via binding of PD-L2), thus enhancing the cytotoxic effect of antitumor T cells.[11]
- It is hypothesized that the combination of a PD-1 receptor antibody with *Lm*-LLO immunotherapy that generates PSA-specific cytotoxic T cells could benefit patients with diseases where PD-1 therapy has not previously exhibited activity.
- This hypothesis is supported by the results of several preclinical studies in mouse tumor models in which antibodies against PD-1 or PD-L1 demonstrated antitumor activity.[12-15]
 - Hence, combination therapy with an *Lm*-LLO immunotherapy targeting PSA and a PD-1 inhibitor has the potential to effectively treat patients with PC
- The present phase 1/2 trial (NCT02325557) has been initiated to determine the safety, tolerability, and efficacy of ADXS31-142 administered as monotherapy and in combination with pembrolizumab to patients with mCRPC.

Figure 1. Step-by-step *Lm*-LLO immunomodulation

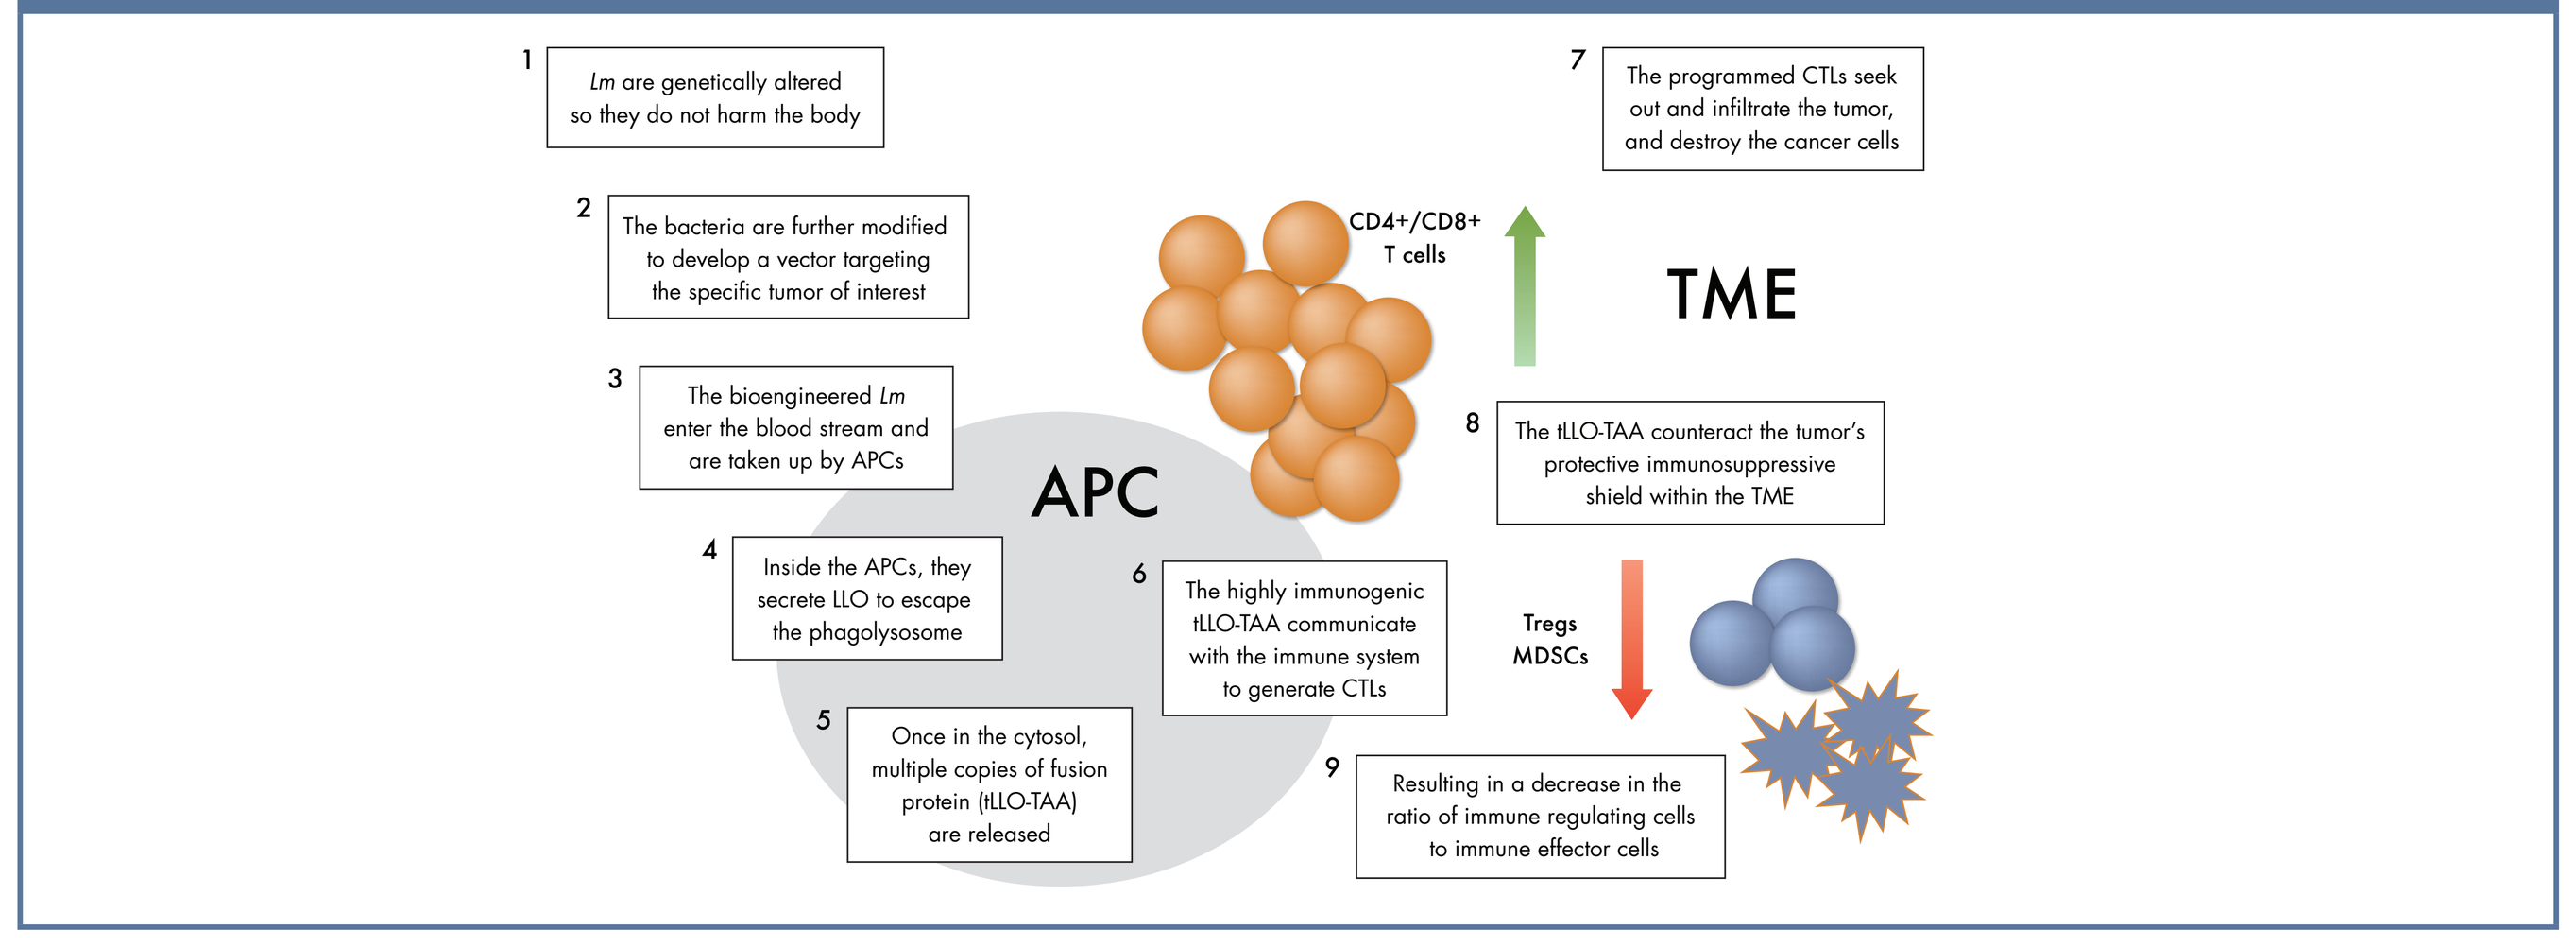
1 Lm are genetically altered so they do not harm the body
2 The bacteria are further modified to develop a vector targeting the specific tumor of interest
3 The bioengineered Lm enter the blood stream and are taken up by APCs
4 Inside the APCs, they secrete LLO to escape the phagolysosome
5 Once in the cytosol, multiple copies of fusion protein (tLLO-TAA) are released
6 The highly immunogenic tLLO-TAA communicate with the immune system to generate CTLs
7 The programmed CTLs seek out and infiltrate the tumor, and destroy the cancer cells
8 The tLLO-TAA counteract the tumor's protective immunosuppressive shield within the TME
9 Resulting in a decrease in the ratio of immune regulating cells to immune effector cells
APC
TME
CD4+/CD8+ T cells
Tregs MDSCs

APC, antigen-presenting cell; CTL, cytotoxic T lymphocyte; LLO, listeriolysin O; *Lm*, *Listeria monocytogenes*; MDSCs, myeloid-derived suppressor cells; TAA, tumor-associated antigen; tLLO, truncated LLO; TME, tumor microenvironment; Tregs, regulatory T cells.

OBJECTIVES

- The study objectives are outlined in **Table 1**.

Table 1. Study objectives

	Part A (Phase 1) ADXS31-142	Part B (Phase 1/2) ADXS31-142 + pembrolizumab
Primary objective(s)	• Evaluate safety and tolerability of ADXS31-142 • Determine the RP2D of ADXS31-142 for Part B	• Evaluate safety and tolerability of ADXS31-142 + pembrolizumab • Determine the RP2D of the combination of ADXS31-142 + pembrolizumab
Secondary objective	• Evaluate antitumor activity and PFS of ADXS31-142 monotherapy and ADXS31-142 + pembrolizumab combination therapy using RECIST v1.1, irRECIST, and PCWG2 criteria	
Exploratory objective	• Evaluate associations between biomarkers of immunologic response (serum PSA) with tumor response and PFS of ADXS31-142 monotherapy and ADXS31-142 + pembrolizumab combination therapy	

irRECIST, immune-related Response Evaluation Criteria In Solid Tumors; PFS, progression-free survival; PCWG2, Prostate Cancer Working Group 2; PSA, prostate-specific antigen; RECIST, Response Evaluation Criteria In Solid Tumors; RP2D, recommended phase 2 dose.

METHODS

STUDY DESIGN

- The study is divided into two parts: Part A (phase 1 design) and Part B (phase 1/2 design) **(Figure 2)**.

Figure 2. Study design

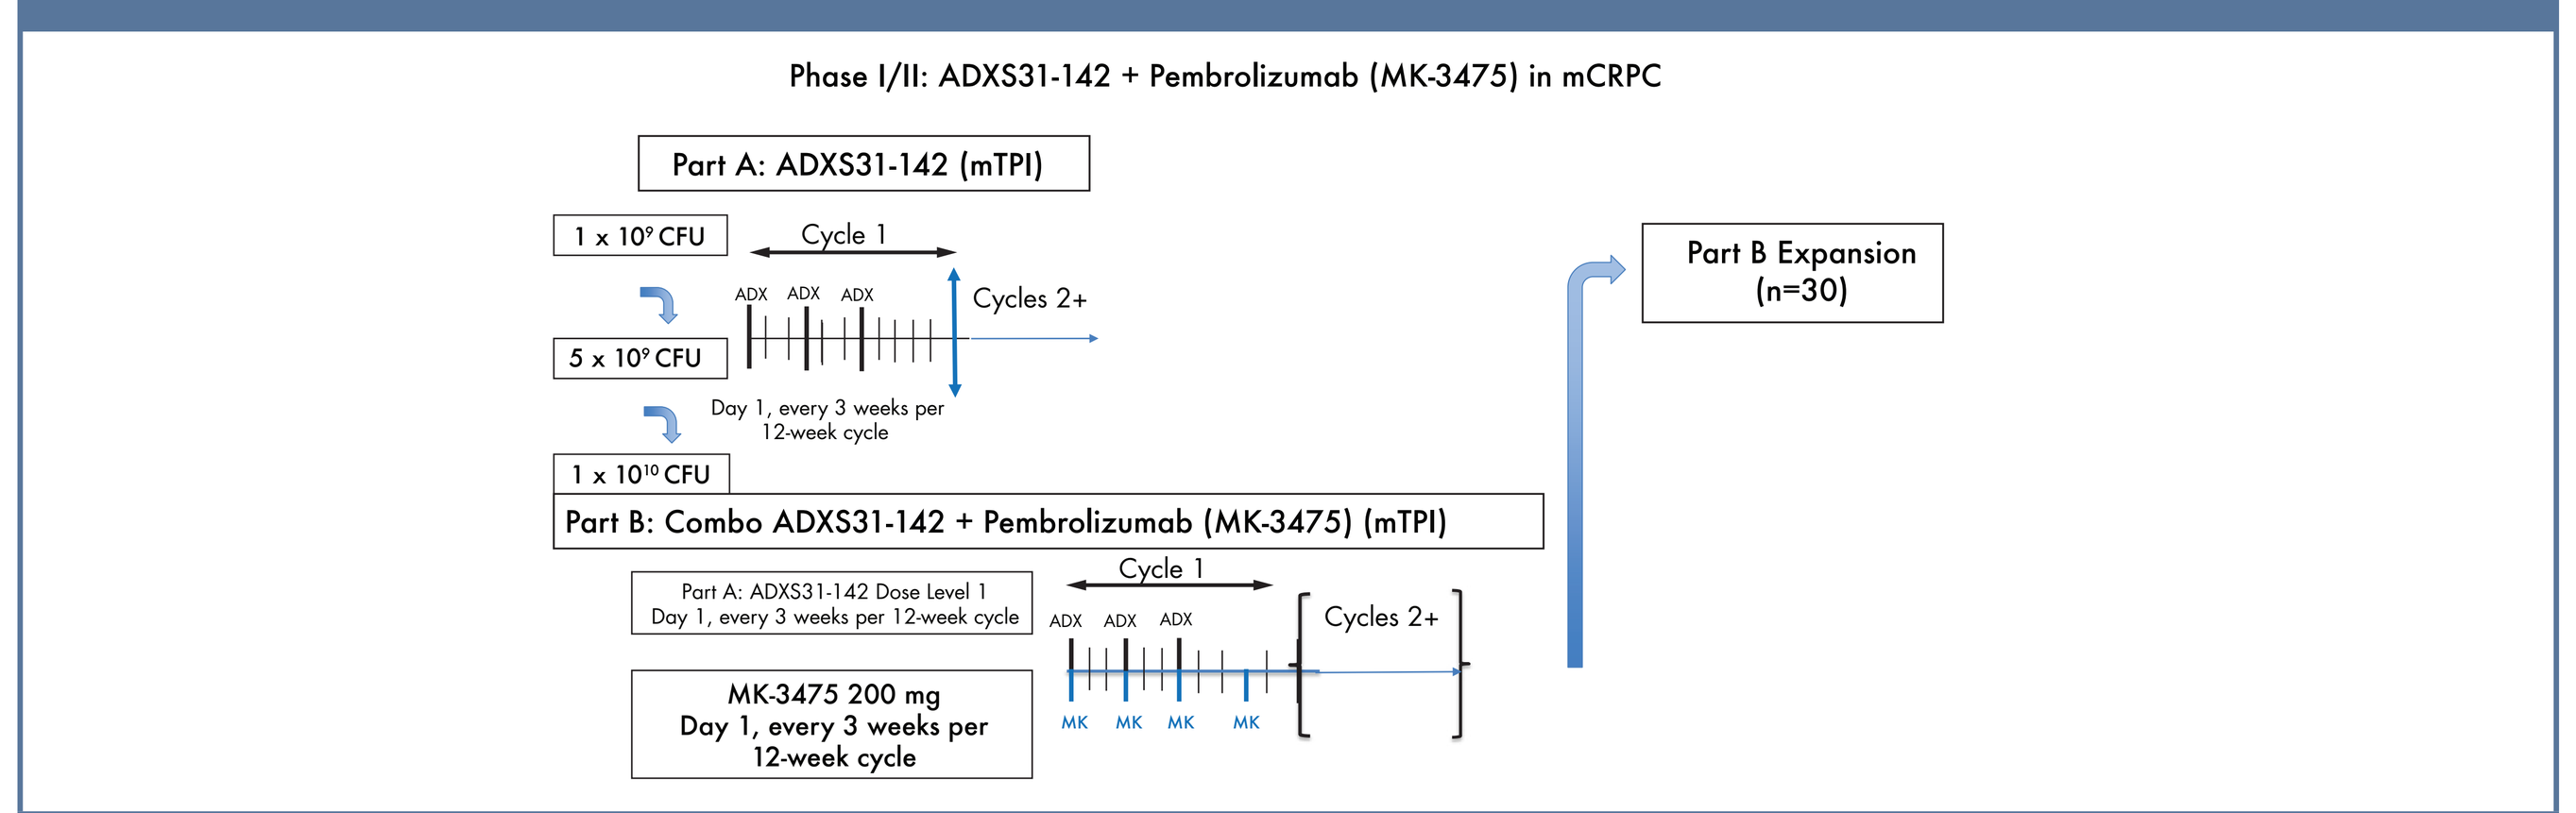
Phase I/II: ADXS31-142 + Pembrolizumab (MK-3475) in mCRPC
Part A: ADXS31-142 (mTPI)
1 x 10⁹ CFU
5 x 10⁹ CFU
1 x 10¹⁰ CFU
Cycle 1
Cycles 2+
ADX ADX ADX
Day 1, every 3 weeks per 12-week cycle
Part B: Combo ADXS31-142 + Pembrolizumab (MK-3475) (mTPI)
Part A: ADXS31-142 Dose Level 1 Day 1, every 3 weeks per 12-week cycle
MK-3475 200 mg Day 1, every 3 weeks per 12-week cycle
MK MK MK MK
Part B Expansion (n=30)

ADX, ADXS31-142; CFU, colony-forming units; mCRPC, metastatic castrate resistant prostate cancer; MK, MK-3475; mTPI, modified toxicity probability interval.

- Part A:
 - Phase 1, open-label, multicenter, nonrandomized, dose-determining trial of ADXS31-142 monotherapy in patients with mCRPC
 - The starting dose (Dose Level 1) for ADXS31-142 will be 1×10^9 colony-forming units (CFU) every 3 weeks (Q3W) for 3 doses in a 12-week cycle
 - Dose will be escalated (5×10^9 CFU, 1×10^{10} CFU), remain the same, or be de-escalated based on the predefined dose-limiting toxicity (DLT) criteria in accordance with the modified toxicity probability interval (mTPI) design (see **Figure 3**)
 - Compared with the 3 + 3 design, the mTPI method exposes fewer patients to toxic doses above the maximum tolerated dose (MTD) and yields similar probabilities in identifying the correct MTD even with matched sample sizes[16]
 - The targeted DLT rate for monotherapy will be ≤25%
 - Up to 21 patients will be enrolled with a minimum of 6 patients treated at the recommended phase II dose (RP2D) before proceeding to the next phase
- Part B:
 - Phase 1/2, open-label, multicenter, nonrandomized, dose-determining trial of ADXS31-142 plus pembrolizumab in patients with mCRPC. This dose-determining phase will be followed by an expansion cohort phase
 - The dose-determining phase will identify a RP2D for the combination therapy; dose escalation/de-escalation will be examined using the mTPI design
 - The starting dose for ADXS31-142 will be one level below the RP2D established for monotherapy, or 1×10^9 CFU, if this dose was the MTD for monotherapy
 - The dosing frequency will remain the same as it is in Part A: Q3W for three doses in a 12-week cycle
 - ADXS31-142 doses will be escalated, remain the same, or be de-escalated based on the predefined DLT criteria in accordance with the mTPI design **(Table 2)**
 - The dose for pembrolizumab will remain fixed at 200 mg and will be administered at the same frequency as ADXS31-142
 - The targeted DLT rate for combination therapy will be ≤30%
 - Either the MTD or the maximum allowable dose for the combination will be the RP2D for the dose-expansion cohort
 - The expansion cohort will enroll up to 30 patients at the RP2D determined in the dose-determination phase to further assess the safety and clinical activity of combination therapy
 - Adverse events (AEs) will be monitored throughout the study and graded according to the National Cancer Institute's Common Terminology Criteria for Adverse Events version 4.0 (NCI CTCAE v4.0)
- In both parts/phases:
 - Patients will receive monotherapy or combination therapy until one of the following occurs: documented disease progression, unacceptable AE(s), intercurrent illness that prevents further administration of treatment, investigator's decision to withdraw the patient, patient withdraws consent, patient experiences a complete response and receives one additional cycle of treatment, noncompliance with trial treatment or procedure requirements, completion of 24 months of treatment with ADXS31-142 and pembrolizumab, or administrative reasons
 - All patients will be monitored for a minimum period of 30 days for AEs or 90 days for serious AEs or events of clinical interest (ECIs)

Figure 3. mTPI decision tables for selection of dose for Part A and Part B of the study

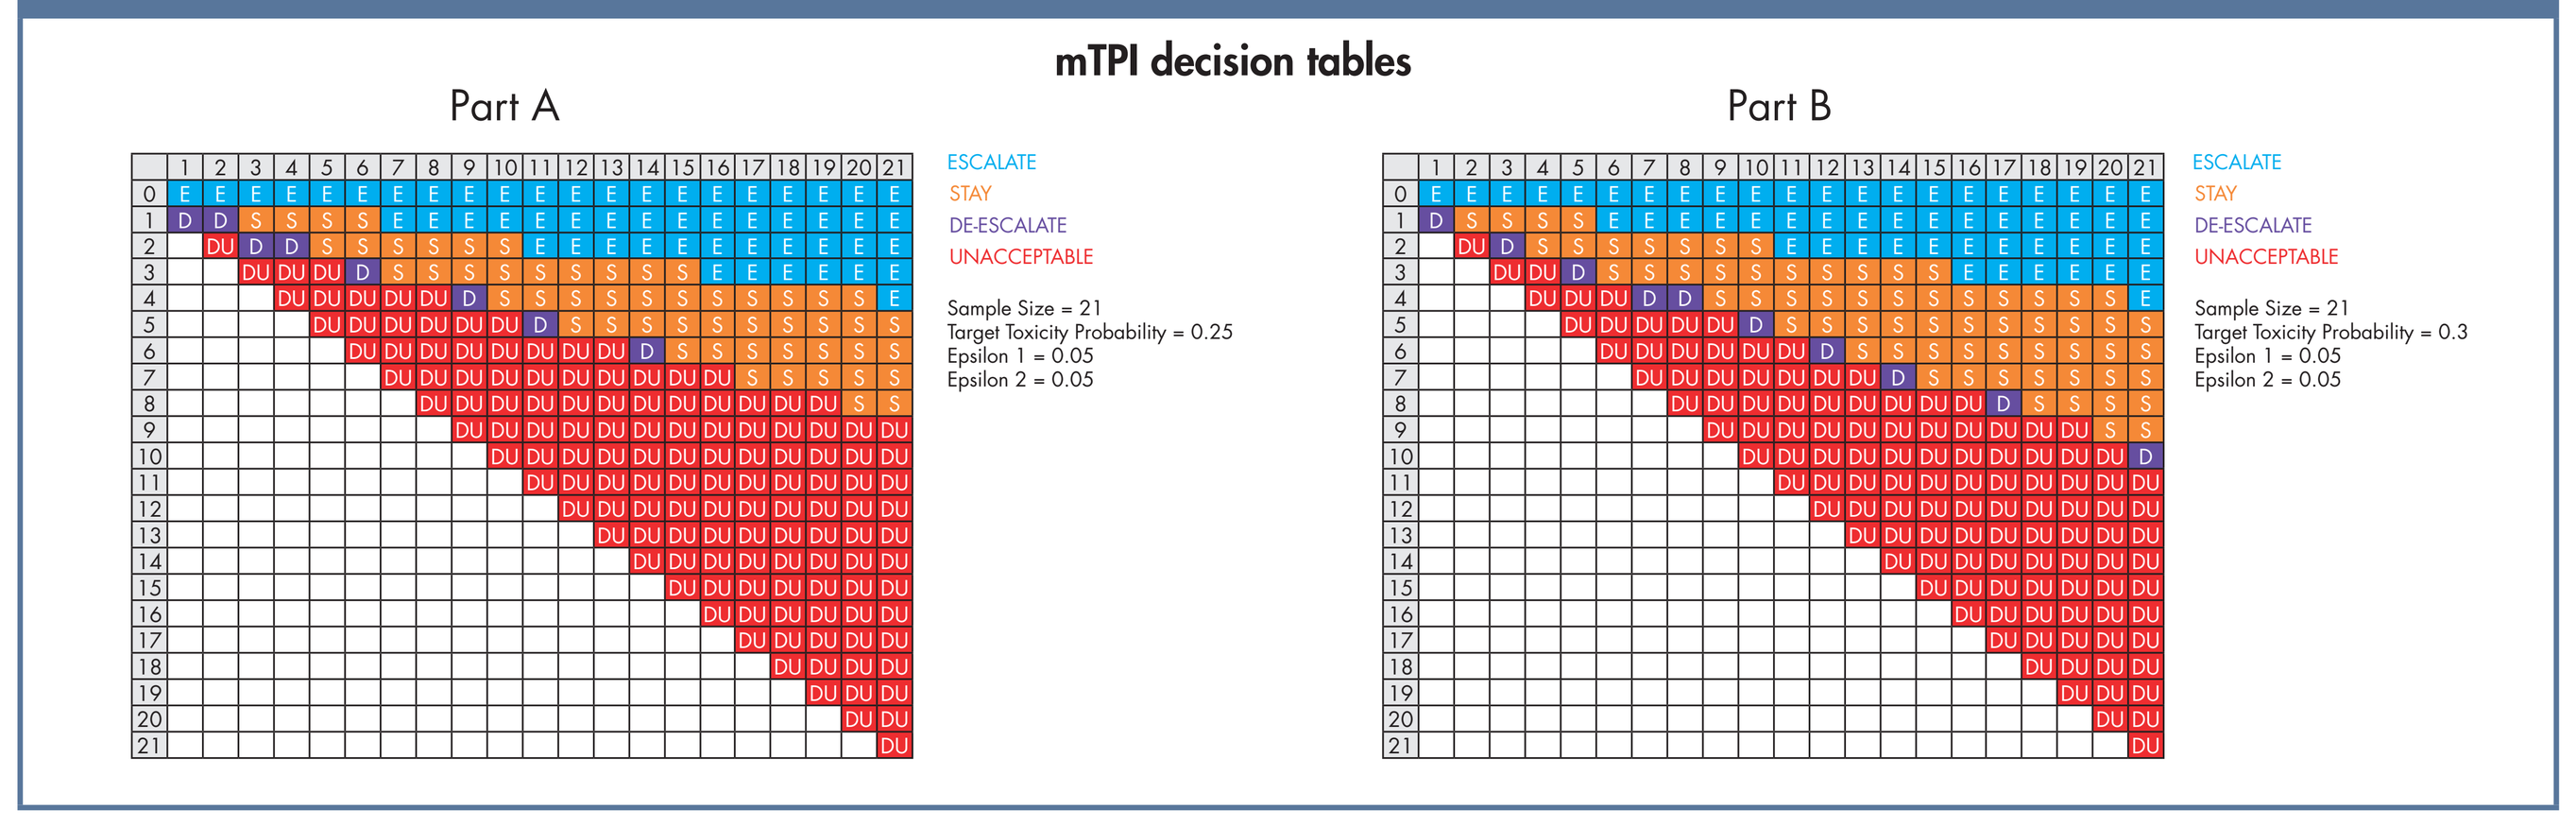
mTPI decision tables
Part A
Part B
ESCALATE
STAY
DE-ESCALATE
UNACCEPTABLE
Sample Size = 21
Target Toxicity Probability = 0.25
Epsilon 1 = 0.05
Epsilon 2 = 0.05
Sample Size = 21
Target Toxicity Probability = 0.3
Epsilon 1 = 0.05
Epsilon 2 = 0.05

mTPI, modified toxicity probability interval.

DOSE-LIMITING TOXICITIES

- DLTs will be graded in both parts of the study using NCI CTCAE v 4.0 after administration of two doses of each drug (period of four weeks).
- The occurrence of any of the toxicities outlined in **Table 2** will be considered a DLT if judged by the investigator to be possibly, probably, or definitely related to the combination therapy.

Table 2. Dose-limiting toxicities

Toxicity categories	Criteria for dose-limiting toxicity
General	Prolonged delay (>2 weeks) in initiating the second dose due to treatment-related toxicity
	Missing the second dose of ADXS31-142 or pembrolizumab as a result of AEs during the first cycle
Hematologic	Grade 4 hematologic toxicity
	Febrile neutropenia (defined as ANC <1000/mm³ at a single temperature of >38.3° C [101° F] or a sustained temperature of >38° C [100.4° F] lasting longer than 1 hour)
	Grade 3 thrombocytopenia lasting >72 hours
	Grade 4 thrombocytopenia
Nonhematologic	≥ Grade 3 nonhematologic toxicity (excluding nausea, vomiting, and/or diarrhea lasting <3 days and reversible with medical intervention)
	Grade 3 nonhematologic laboratory value that does not downgrade to grade 2 within 3 days after onset despite optimal medical management
	Listeremia: persistent (for 48–72 hours postdose) symptoms consistent with listeremia (eg, fever and muscle aches, often preceded by diarrhea or other gastrointestinal symptoms)
	Grade 3 cytokine release symptoms lasting >24 hours after ADXS31-142 administration despite symptomatic treatment

AEs, adverse events; ANC, absolute neutrophil count.

INTERVENTIONS

- In Part A, patients will receive ADXS31-142 on day 1 of week 1, 4, and 7, and in Part B patients will receive ADXS31-142 at the same frequency along with pembrolizumab Q3W in each 12-week treatment cycle.
- ADXS31-142 will be administered as a 60-minute intravenous (IV) infusion, while pembrolizumab will be administered as a 30-minute IV infusion.
- Nonsteroidal anti-inflammatory drugs (NSAIDs), antihistamines, and antiemetics will be administered to all patients 30 minutes prior to the ADXS31-142 infusion.
- In Part B, there should be approximately 60 minutes between the end of the first infusion and the start of the second infusion. Patients will receive the pembrolizumab infusion first, followed by administration of NSAIDs and oral antiemetics within 30 minutes prior to the ADXS31-142 infusion.
- Patients receiving ADXS31-142 will receive antibiotics 72 hours after each ADXS31-142 infusion.
- If needed, patients will be administered additional doses of NSAIDs (approximately every four hours) and antiemetics (approximately every eight hours) following the ADXS31-142 infusion.

PATIENT ELIGIBILITY

- Key patient eligibility criteria are described in **Table 3**.

Table 3. Key patient eligibility criteria

Key inclusion criteria
Adult male patients (≥18 years)
Progressive mCRPC on androgen deprivation therapy identified by either • PSA progression of 25% increase over baseline (increase in absolute value of at least 2 ng/mL) confirmed by a second measure within a 1-week interval OR • Progression of nodal metastasis assessed within 1 month prior to screening with CT or MRI of abdomen and pelvis OR • Progression of evaluable bone disease
Discontinued antiandrogens (bicalutamide, nilutamide) >6 weeks and enzalutamide >4 weeks prior to day 1 of trial treatment
ECOG PS ≤1
Adequate hematologic, hepatic, and renal function
Key exclusion criteria
Received >3 prior systemic treatment regimens with chemotherapy, hormonal, or immunotherapy in the metastatic setting or received more than 1 prior chemotherapeutic regimen in the metastatic setting
Received prior chemotherapy, targeted small-molecule therapy, or radiation therapy within 2 weeks prior to study day 1 or has not recovered (ie, ≤ grade 1 or at baseline) from AEs
Has known active central nervous system metastases and/or carcinomatous meningitis or an additional malignancy that is progressing or requires active treatment (except basal cell carcinoma/squamous cell carcinoma of the skin or superficial urothelial cancer 2 years post-therapy)
Diagnosed with immunodeficiency or received systemic steroid therapy, abiraterone acetate, or other androgen synthesis inhibitors or immunosuppressive therapy within 7 days, or a monoclonal antibody within 4 weeks prior to study day 1, or a live vaccine within 30 days of first trial dose
Has an active autoimmune disease requiring systemic treatment within the past 2 years or evidence of interstitial lung disease, or active noninfectious pneumonitis
Known contraindication to study antibiotics or NSAIDs and allergy to any component of the study drug(s) formulations
Known history of listeriosis, human immunodeficiency virus, and/or known active hepatitis B or C
Has an active infection requiring systemic therapy

AEs, adverse events; CT, computed tomography; CTCAE, Common Terminology Criteria for Adverse Events; ECOG PS, Eastern Cooperative Oncology Group performance status; mCRPC, metastatic castration-resistant prostate cancer; MRI, magnetic resonance imaging; NSAIDs, nonsteroidal anti-inflammatory drugs; PSA, prostate-specific antigen.

ENDPOINTS

- Safety:
 - Safety will be assessed by grading AE severity as per NCI CTCAE v3.0 for treatment and as per NCI CTCAE v4.0 for concomitant medications. Endpoints will also include physical examinations, vital signs, biochemical parameters, DLTs, serious AEs, ECIs, and irECIs experienced by patients who have received ADXS31-142 alone or in combination with pembrolizumab
- Efficacy:
 - Serum levels of biomarkers such as PSA/PAP and other serum markers of PC, PSA doubling time, and computerized tomography (CT) and magnetic resonance imaging (MRI) scans will be used to assess antitumor activity
 - Measurable and evaluable disease assessments such as tumor response, time to tumor progression (progression-free survival [PFS] up to 24 months), and survival up to 24 months will also be used to assess efficacy
 - Pharmacokinetic and antidrug antibody determinations will be assessed from blood samples in the pembrolizumab arm
- Immunologic:
 - T cells will be assessed for their specific response to PSA, PAP, and others using enzyme-linked immunosorbent assay (ELISA) and/or ELISpot
 - Serum cytokine and chemokine changes will also be assessed for immune stimulation as a function of treatment

STATISTICAL METHODS

- Descriptive statistics will be used to summarize and evaluate the safety and tolerability of ADXS31-142 alone and in combination with pembrolizumab.
- All patients who received at least one dose of ADXS31-142 or pembrolizumab will be included in the safety analyses, while those patients who complete at least one cycle of treatment will be included in the efficacy analyses.
- Safety data will be summarized by evaluating physical examinations, serum chemistry values, and hematology values and compared to baseline or historical controls.
- Efficacy will be assessed by evaluating changes in PSA and tumor response, with data from CT/MRI scans and use of RECIST criteria, irRECIST criteria, and overall survival compared to historical controls.
- Immunology will be assessed by comparing pre- and posttreatment cytokine values, flow cytometric analysis of peripheral blood mononuclear cells, and the response of T cells to antigen-specific stimulation. Correlations will be made between immunologic responses and clinical responses.

TRIAL STATUS

- Five of the nine trial centers in the United States are recruiting patients for the study, and as of October 2015, five patients have been enrolled.

REFERENCES

1. National Cancer Institute. Surveillance, Epidemiology, and End Results Program. 2015. http://seer.cancer.gov/statfacts/html/prost.html. Accessed October 12, 2015.
2. Daverede L, et al. J Med Case Rep 2014;8:122.
3. Kloor M. Lancet Oncol 2009;10:840-41.
4. Kantoff PW, et al. N Engl J Med 2010;363:411-22.
5. Gunn GR, et al. J Immunol 2001;167:6471-9.
6. Maciag PC, et al. Cancer Res 2008;68:8066-75.
7. Singh R, et al. J Immunol 2012;188:165.12.
8. Hussain SF & Paterson Y. J Immunother 2004;27:339-46.
9. Advaxis Immunotherapies. Advaxis Submits Investigational New Drug Application for ADXS31-142 (ADXS-PSA) for the Treatment of Metastatic Castration Resistant Prostate Cancer (press release). http://ir.advaxis.com/press-releases/detail/854. Accessed October 12, 2015.
10. Patnaik A, et al. Clin Cancer Res 2015;21:4286-93.
11. Hiraoka N. Int J Clin Oncol 2010;15:544-51.
12. Hirano F, et al. Cancer Res 2005;65:1089-96.
13. Iwai Y, et al. Proc Natl Acad Sci U S A 2002;99:12293-7.
14. Okudaira K, et al. Int J Oncol 2009;35:741-9.
15. Zhang C, et al. Cytotherapy 2008;10:711-9.
16. Ji Y & Wang SJ. J Clin Oncol 2013;31:1785-91.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster.
http://bit.ly/sitc-2015-advaxis-posters

ACKNOWLEDGMENTS

Editorial and medical writing assistance was provided by Siddharth Mukherjee, PhD, TRM Oncology, The Netherlands, funded by Advaxis. The authors are fully responsible for all content and editorial decisions for this poster.

DISCLOSURES

Advaxis Disclosures: Advaxis, Inc. and Merck & Co., Inc. provided financial support for the study and participated in the design, study conduct, analysis and interpretation of data, as well as the writing, review, and approval of the poster. ADXS31-142 and pembrolizumab are being developed by Advaxis, Inc. and Merck & Co., Inc respectively.

Naomi B. Haas, Mark N. Stein, Ronald Tutrone: No disclosures. Rodolfo Perini, Andrew Denker: Full-time employee of Merck and Co. Inc. David Mauro: Employee and stakeholder of Advaxis, Inc. Lawrence Fong: Research support from Advaxis.